Population pharmacokinetic modelling of CHF6001 following dry powder inhalation in healthy volunteers

Koen Jolling (1), Massimo Cella (2), Mirco Govoni (2), Anna Nandeuil (3), Andreas Lindauer (1)

[1] SGS Exprimo, Mechelen, Belgium; [2] Chiesi Farmaceutici S.p.A., Via Palermo 26/A, 43122 Parma, Italy;
[3] Chiesi Farmaceutici S.p.A., Paris, France

Background & objectives

CHF6001 is a potent and selective phosphodiesterase-4 (PDE-4) inhibitor to treat chronic obstructive pulmonary disease (COPD) and asthma. CHF6001 is being developed for inhalation to help overcome the well-known gastrointestinal side effects associated with this therapeutic class when given orally. While initially available in Hard Gelatine Capsules delivered by the Aerolizer® device, CHF6001 is now developed via the novel multi-dose NEXThaler® inhaler. The objectives of the analysis were:

- to evaluate the population pharmacokinetics (PK) of CHF6001 and the influence of selected covariates in healthy volunteers
- to simulate CHF6001 plasma profiles of relevant clinical doses to be administered via the NEXThaler® inhaler

Methods

Patients & Data: Data from 2 phase I (FIH and Extension) double-blind, randomized, placebo-controlled studies with single (SAD) and repeated ascending doses (MAD).

- **FIH**: 61 subjects with once daily administration via the Aerolizer® inhaler (20 - 2000 µg).
- **Extension**: 39 subjects with twice daily administration via the NEXThaler® inhaler (2400 - 4800 µg)

In total, 2931 CHF6001 plasma concentrations, collected in 100 healthy volunteers, were included.

Model development and parameter estimations were performed with NONMEM 7.3.

Model Characteristics:

- Population approach, accounting for inter-individual variability (IIV) on PK parameters
- Study specific IIV and inter-occasion variability (IOV) were examined: single/multiple dose and at trough

Covariate selection:

- Explored covariates: *FORM (NEXThaler® or Aerolizer® device), age, sex, body weight, body mass index*
- Forward selection - backward deletion procedure

Simulations:

- Total daily dose of 2400µg: QD and BID with the NEXThaler® device

Results

A two-compartment disposition model, with 3 parallel absorption pathways (slow, intermediate and fast) and first-order elimination was developed (Figure 1), similar to the one proposed by Borghardt *et al.* [1]. For both devices the majority of the available dose, 62.8% and 41.3% of the bioavailable dose for NEXThaler® and Aerolizer®, respectively, was absorbed via the slow pathway (D2, KA2). The smallest available fraction, 10% for NEXThaler® and 12% for Aerolizer®, was absorbed via an early very fast pathway (D3). The remaining fraction, 27.2% for NEXThaler® and 26% for Aerolizer®, was absorbed via the early intermediate pathway (KA1). The absorption rate constant for the latter pathway (KA1) was estimated to be 39.9% higher for Aerolizer® as compared to NEXThaler®.

Figure 1: Scheme of the pop PK model for CHF6001.

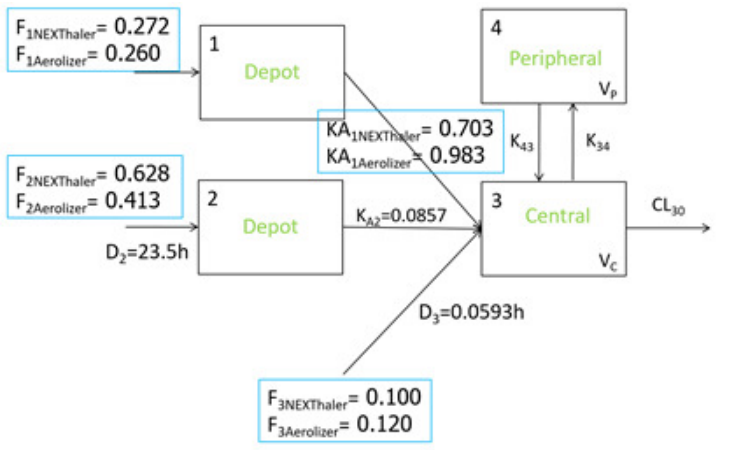
F1NEXThaler= 0.272
F1Aerolizer= 0.260
1
Depot
4
Peripheral
VP
KA1NEXThaler= 0.703
KA1Aerolizer= 0.983
K43
K34
F2NEXThaler= 0.628
F2Aerolizer= 0.413
2
Depot
KA2=0.0857
3
Central
VC
CL30
D2=23.5h
D3=0.0593h
F3NEXThaler= 0.100
F3Aerolizer= 0.120

When using the Aerolizer® device, the median C_{max} was found to be 8% higher, for a dose of 2400 µg, as compared to the NEXThaler® device, while the AUC was found to be 20.7% lower. Simulating the same total daily dose but with different regimens (i.e. QD vs. BID) via the Nexthaler® device, a similar 24h exposure was obtained, but with BID dosing resulting in 35% lower fluctuation (calculated as C_{max}-C_{min}/C_{av}) and 11% lower Cmax. Parameter estimates are reported in Table 1, simulations in Figure 2 and visual predictive checks stratified by device in Figure 3.

Figure 2: Simulated CHF6001 profiles after single and multiple administrations of 2400 µg bid

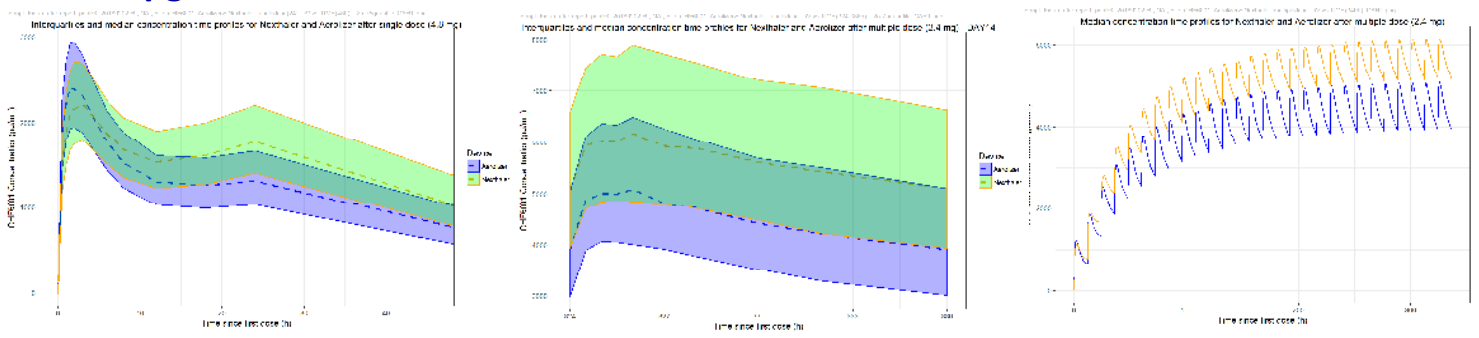

Table 1: Estimates of model parameters (typical values)

Parameter	Estimate	Bootstrap[a] Median	Bootstrap[a] 90% CI	Shr.(%)[c]
Fixed effects parameters				
CL/F (L/h)	34.8	34.3	31 - 38.1	-
V3/F (L)	537	526	434 - 605	-
Q/F (L/h)	47.3	49.6	32 - 99.6	-
V4/F (L)	977	1004	772 - 1423	-
KA1 NEXThaler (1/h)	0.704	0.689	0.508 - 0.832	-
KA2 (1/h)	0.0857	0.0871	0.0612 - 0.284	-
D2 (h)	23.6	23.7	22.9 - 25.8	-
D3 (h)	0.0585	0.0597	0.0566 - 0.084	-
PBIO: Relative bioavailability NEXThaler	1 (fixed)	-	-	-
FF1: Available fraction NEXThaler	0.272	0.277	0.237 - 0.334	-
FF2: Available fraction NEXThaler	0.862	0.864	0.834 - 0.894	-
PBIO: Relative bioavailability Aerolizer	0.793	0.788	0.708 - 0.891	-
FF2: Available fraction Aerolizer	0.774	0.773	0.729 - 0.826	-
KA1 Aerolizer (change as compared to NEXThaler)	0.399	0.378	0.163 - 0.675	-
FF1: Available fraction Aerolizer	0.328	0.334	0.278 - 0.395	-
Random effects parameters				
IIV_CL/F (CV%)[b]	29	28.9	23 - 34.1	17.9
IIV_V3/F (CV%)[b]	15.9	16	3.59 - 24.4	49.5
IIV_V4/F (CV%)[b]	77.3	76.3	48.1 - 110	21.5
IIV_KA1 (CV%)[b]	26.3	24	0.222 - 35	40.2
IIV_KA2 (CV%)[b]	30.3	29	7.49 - 118	42.7
IIV_Q/F (CV%)[b]	32	29.1	0.378 - 53.9	40.2
IIV_PBIO (CV%)[b]	20.3	20.1	15.8 - 23.6	22.2
IIV_FF1 (CV%)[b]	26.3	26	15.1 - 36.4	30
IIV_FF2 (CV%)[b]	47.6	46.4	33.9 - 57.6	25.7
Residual variability				
Residual error proportional (sd/mean)	0.114	0.114	0.108 - 0.121	7.1
Residual error additive (pg/mL)	12.7	12.2	6.26 - 18	-

CV: Coefficient of variation, CL/F: Apparent Clearance, V3: Apparent Central volume of distribution, V4: Apparent peripheral volume of distribution, KA: Absorption rate constant, D: duration, IIV: inter-individual variability

[a] 500 runs out of 500

[b] CV = 100*sqrt(exp(variance)-1)

[c] Epsilon-shrinkage is the overall shrinkage for related elements of the residual error

Figure 3: Visual Predictive Checks of CHF6001 plasma concentrations as a function of time since first dose

DAY1 (left panel), DAY7 (right upper panel) and DAY14 (right lower panel) **Trough values**

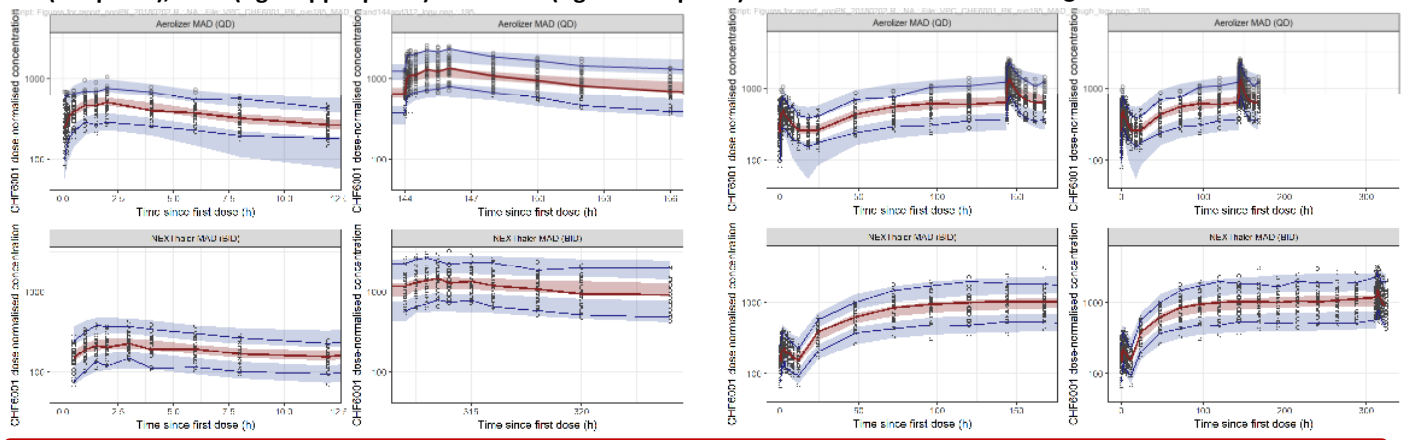
Aerolizer MAD (QD)
NEXThaler MAD (BID)
Time since first dose (h)
CHF6001 dose-normalised concentration

Conclusions

A PK model with 3 parallel absorption pathways described the CHF6001 concentration data very well and was able to capture a second peak observed 24h after dosing. With NEXThaler® more drug substance is delivered for a given nominal amount compared to the Aerolizer® device.

[1] Borghardt *et al*. Investigating pulmonary and systemic pharmacokinetics of inhaled olodaterol in healthy volunteers using a population pharmacokinetic approach. Br J Clin Pharmacol. 2016 Mar;81(3):538-52