Poster IV-087 – Abstr. 11079

How to handle population pharmacokinetics modeling in the context of a regulatory submission for a compound already well-characterized in other settings? A case-study with nivolumab in previously untreated urothelial cancer.

Mélanie Wilbaux,[1] Yue Zhao,[2] Anna Kondic,[2] Lora Hamuro,[2] Amit Roy[1]
[1]Pumas-AI Inc.; [2]Bristol Myers Squibb, Princeton, NJ, USA

Introduction

- **PopPK Modeling in Drug Development:**
 - Routinely used to characterize the pharmacokinetics (PK) of a compound throughout its clinical development lifecycle
 - Enables characterization of exposure-response (E-R) of efficacy, safety, and biomarkers in the target patient population to inform dose selection and justification
- **Extending the Use of a Drug to a New Setting (Patient Population + Treatment):**
 - Decisions regarding dosing strategies for a new setting is informed by clinical trial data and PK/PD considerations.
 - Characterizing PK for subsequent indication facilitates the bridging of safety data from previous indications to the new one and enables characterization of E-R relationships.
 - → In such scenario, the following question emerges: Has the initial PopPK model been adequately characterized and validated to accurately represent the new clinical data, and can it be utilized for predicting individual exposures?
- **A Case-Study with Nivolumab in Urothelial Cancer:**
 - A PD-1 inhibitor already approved for multiple indications, both as monotherapy and in combination with ipilimumab or chemotherapy [1, 2]
 - Nivolumab PK, after intravenous infusion, has been extensively characterized [3].
 - New Phase 3 data evaluating nivolumab in combination with chemotherapy vs. chemotherapy alone for previously untreated unresectable or metastatic urothelial cancer (UC)

 → To characterize nivolumab PK in UC by leveraging prior characterization of PK in two indications (non-small cell lung cancer (NSCLC) and melanoma (MEL)), and derive exposure for E-R analyses to support regulatory submission for this new indication

Objective: To explore different methodologies of handling PopPK modeling for a compound already well-characterized in other settings (Patient Population + Treatment)

Figure 1. Comprehensive Framework for PopPK Modeling of a Well-characterized Compound in a New Therapeutic Setting

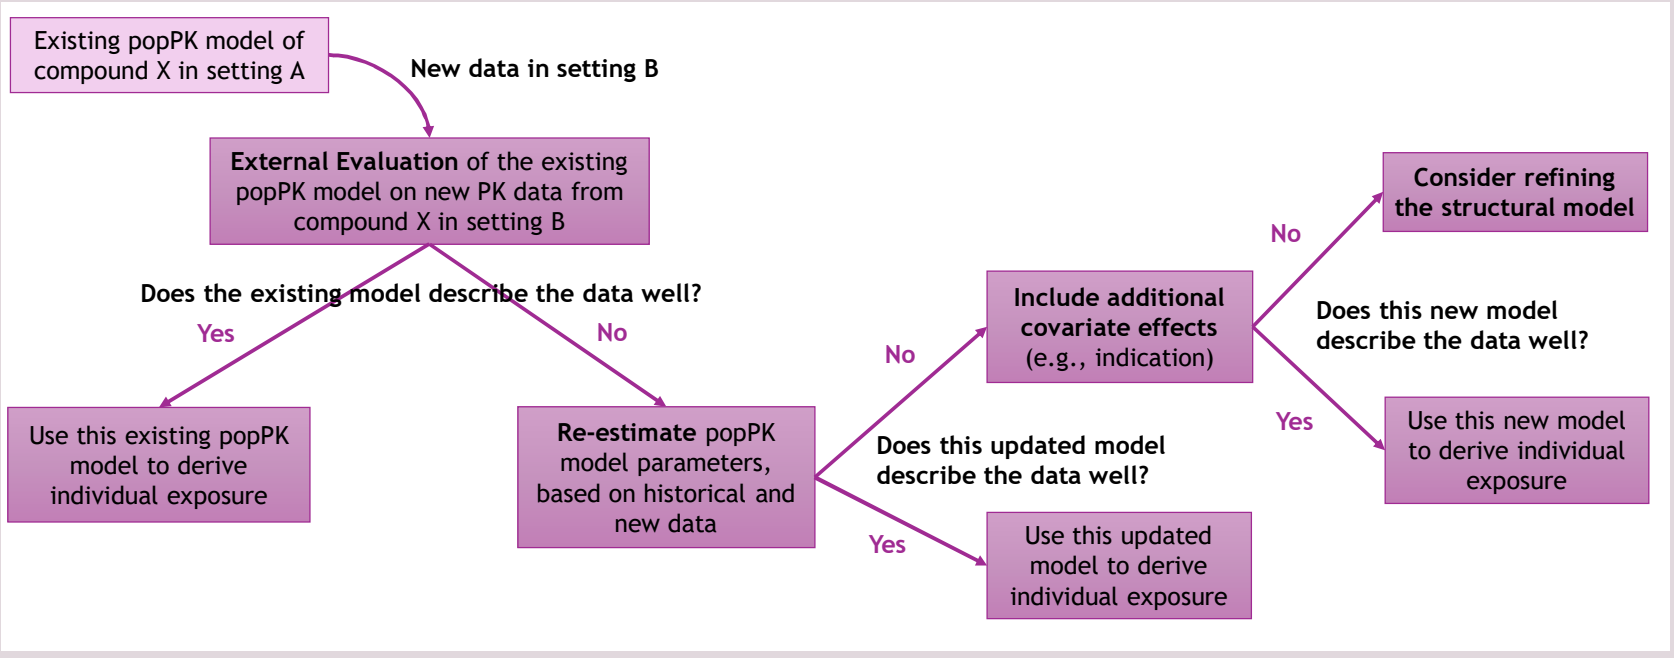
Existing popPK model of compound X in setting A
New data in setting B
External Evaluation of the existing popPK model on new PK data from compound X in setting B
Does the existing model describe the data well?
Yes
No
Use this existing popPK model to derive individual exposure
Re-estimate popPK model parameters, based on historical and new data
Does this updated model describe the data well?
No
Yes
Include additional covariate effects (e.g., indication)
Use this updated model to derive individual exposure
Does this new model describe the data well?
No
Yes
Consider refining the structural model
Use this new model to derive individual exposure

Methods

- The latest developed nivolumab popPK model used data from subjects with previously treated UC, MEL and NSCLC treated with nivolumab monotherapy and in combination with ipilimumab [**Table 1**]
- Nivolumab PK already well-described by a two-compartment model with time-varying clearance (sigmoidal-Emax function)
- Previous characterizations indicate similar clearance across different solid tumor types, lines of therapy and treatment combinations.

Table 1.Historical and New Data from the Analysis Dataset

Tumor Type	Line of Therapy	Nivolumab Monotherapy of Combination	Nivolumab PK
Melanoma	1L and 2L+	Monotherapy	Already described
NSCLC	1L and 2L+	Monotherapy	Already described
UC	2L +	Monotherapy	Already described
UC	1L	Combination with chemotherapy	**Not previously described**

Abbreviations: NSCLC = non-small cell lung cancer; UC = urothelial carcinoma; 1L = first line ; 2L+ = second line and more
Note: Subjects who received combination with ipilimumab were not included in the analysis dataset

Figure 2. Nivolumab PopPK Modeling Strategy in Previously Untreated Urothelial Cancer

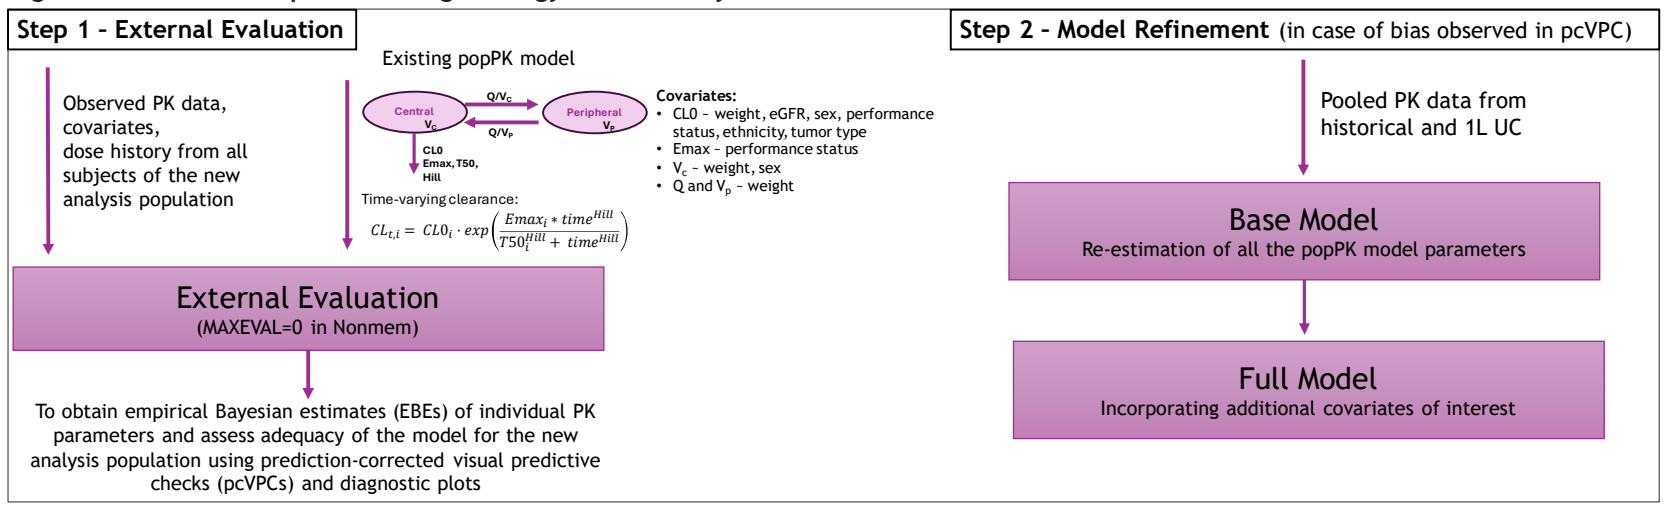
Step 1 - External Evaluation
Existing popPK model
Observed PK data, covariates, dose history from all subjects of the new analysis population
Central V_c
Peripheral V_p
Q/V_c
Q/V_p
CL0
Emax, T50, Hill
Covariates:
CL0 – weight, eGFR, sex, performance status, ethnicity, tumor type
Emax – performance status
V_c – weight, sex
Q and V_p – weight
Time-varying clearance:
$CL_{t,i} = CL0_i \cdot exp\left(\frac{Emax_i * time^{Hill}}{T50_i^{Hill} + time^{Hill}}\right)$
External Evaluation
(MAXEVAL=0 in Nonmem)
To obtain empirical Bayesian estimates (EBEs) of individual PK parameters and assess adequacy of the model for the new analysis population using prediction-corrected visual predictive checks (pcVPCs) and diagnostic plots
Step 2 - Model Refinement (in case of bias observed in pcVPC)
Pooled PK data from historical and 1L UC
Base Model
Re-estimation of all the popPK model parameters
Full Model
Incorporating additional covariates of interest

Results

- **External Evaluation:** external evaluation approach based on pcVPC showed underprediction at the median and 5% percentile and overall inflation of variability [**Figure 3**]
 → The model required to be refined.

Figure 3. External Evaluation

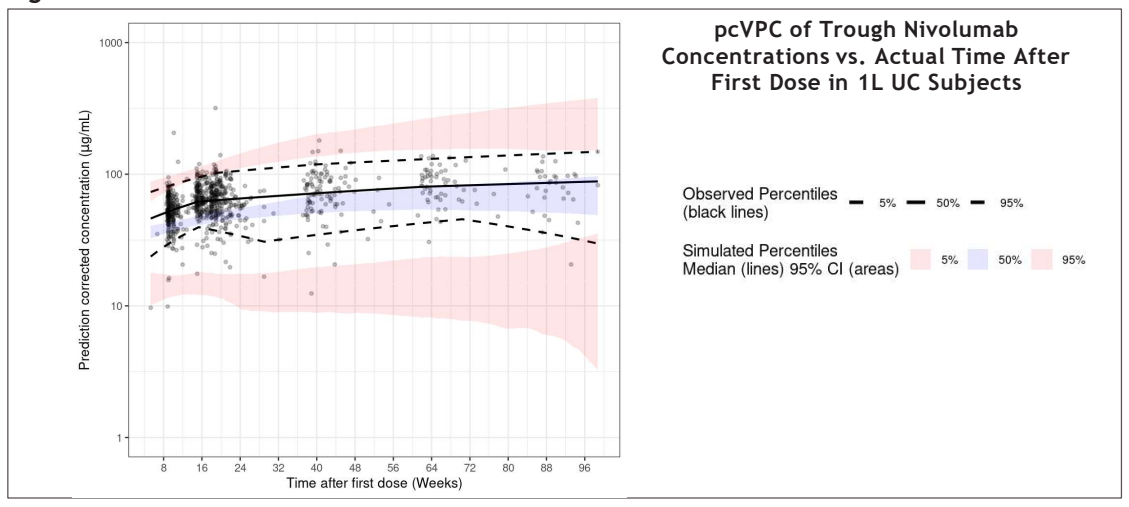
pcVPC of Trough Nivolumab Concentrations vs. Actual Time After First Dose in 1L UC Subjects
Prediction corrected concentration (µg/mL)
Time after first dose (Weeks)
Observed Percentiles (black lines)
5%
50%
95%
Simulated Percentiles Median (lines) 95% CI (areas)
5%
50%
95%

- **Base Model:**
 Re-estimating model parameters showed significant improvement; however remaining underprediction for new data required to investigate additional covariates of interest.
- **Full Model:**
 - Included additional covariate effect:
 - CL0 ~ baseline serum albumin (found to have a significant effect in previous nivolumab popPK models)
 - Emax for time-varying CL ~ combination with chemotherapy (Mono. Vs. Combo)
 - Diagnostic plots and pcVPCs confirmed the appropriateness of the full model [**Figure 4**]

Figure 4. pcVPC from the Full Model Stratified by Population Type

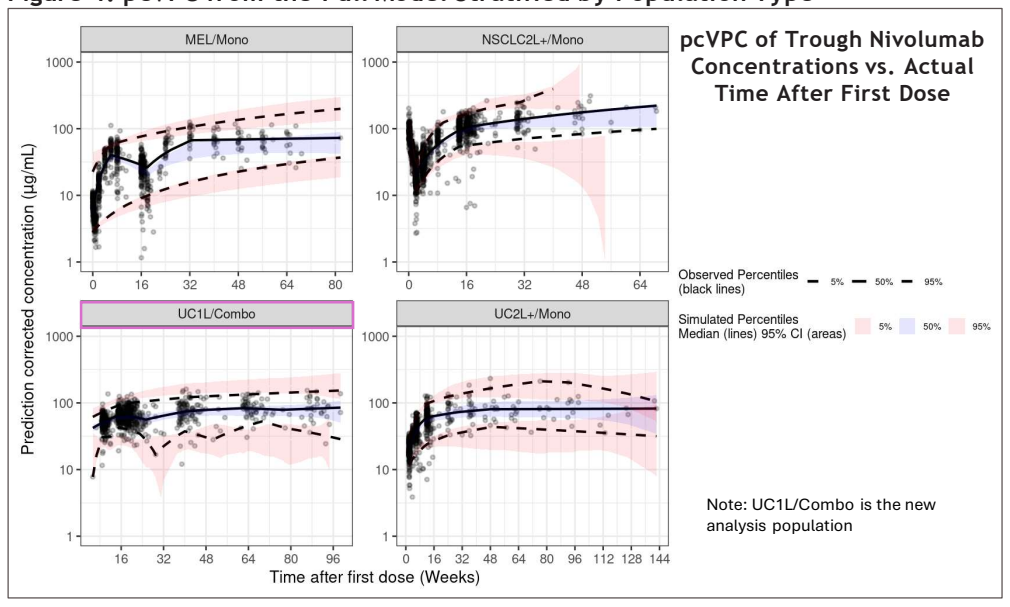
pcVPC of Trough Nivolumab Concentrations vs. Actual Time After First Dose
MEL/Mono
NSCLC2L+/Mono
UC1L/Combo
UC2L+/Mono
Prediction corrected concentration (µg/mL)
Time after first dose (Weeks)
Observed Percentiles (black lines)
5%
50%
95%
Simulated Percentiles Median (lines) 95% CI (areas)
5%
50%
95%
Note: UC1L/Combo is the new analysis population

Abbreviations: Combo = combination with monotherapy; MEL = melanoma; Mono = monotherapy; NSCLC = non-small cell lung cancer; UC = urothelial carcinoma; 1L = first line ; 2L+ = second line and more.

- The model was refined based on the nivolumab PopPK analysis dataset included 6,518 concentration values from 1,355 subjects with MEL, NSCLC, or UC who received nivolumab monotherapy or combination therapy with chemotherapy from 7 clinical studies [**Table 1**].

Table 1. Analysis Population Summary

	MEL Mono N = 106	NSCLC Mono N = 648	UC1L Combo N = 294	UC2L+ Mono N = 307	Total N = 1355
Sex					
Male	71 (67%)	408 (63%)	228 (78%)	236 (77%)	943 (70%)
Female	35 (33%)	240 (37%)	66 (22%)	71 (23%)	412 (30%)
Race					
White	102 (96%)	582 (90%)	203 (69%)	264 (86%)	1151 (85%)
Black/African American	2 (2%)	32 (5%)	0 (0%)	6 (2%)	40 (3%)
Asian	0 (0%)	16 (3%)	74 (25%)	29 (9%)	119 (9%)
Others/Unknown	2 (2%)	18 (2%)	17 (6%)	8 (3%)	45 (3%)
Baseline weight [kg]					
Mean (SD)	83.6 (18.7)	74.4 (16.5)	76.2 (15.8)	78.9 (18.0)	76.6 (17.1)
Median (Min, Max)	81.3 (48.9, 140)	72.7 (34.9, 158)	75.0 (40.6, 136)	79.2 (39.0, 138)	75.0 (34.9, 158)
Baseline eGFR [mL/min/1.73m²]					
Mean (SD)	84.1 (18.6)	83.0 (19.9)	79.5 (17.2)	64.8 (20.4)	78.6 (20.6)
Median (Min, Max)	88.1 (37.4, 131)	85.5 (31.2, 135)	79.7 (43.2, 142)	62.2 (18.1, 117)	80.1 (18.1, 142)
Baseline serum albumin [g/dL]					
Mean (SD)	4.22 (0.492)	3.88 (0.491)	4.02 (0.505)	3.78 (0.487)	3.91 (0.507)
Median (Min, Max)	4.30 (2.60, 5.10)	3.90 (1.90, 5.20)	4.10 (2.40, 5.60)	3.80 (2.20, 5.30)	4.00 (1.90, 5.60)
Baseline performance status					
0	67 (63%)	161 (25%)	159 (54%)	175 (57%)	562 (41%)
1	36 (34%)	483 (74%)	133 (45%)	132 (43%)	784 (58%)
2	3 (3%)	4 (1%)	2 (1%)	0 (0%)	9 (1%)

Abbreviations: MEL = melanoma; NSCLC = non-small cell lung cancer; UC = urothelial carcinoma; 1L = first line ; 2L+ = second line and more.

Discussion

- Differences in PK were due to a combination of factors, including tumor type and combination treatment.
- Tumor type effect is not expected to be clinically relevant: nivolumab CL0 in UC subjects was 7.5% lower than in NSCLC.
- Combination treatment effect is not expected to be clinically relevant: the extent of change in nivolumab CL over time (Emax) in subjects treated with nivolumab in combination with chemotherapy was 6.1 % lower than in those treated with nivolumab a monotherapy.
- Strong effect of baseline serum albumin: nivolumab CL0 was 23% higher at the 5th percentile of baseline serum albumin compared to the reference value.
- The choice of the initial model was critical. Retrospectively, opting for a different model (including chemotherapy effect) might have led to a better fit.

Figure 5. Forest Plot of Key Covariate Effects

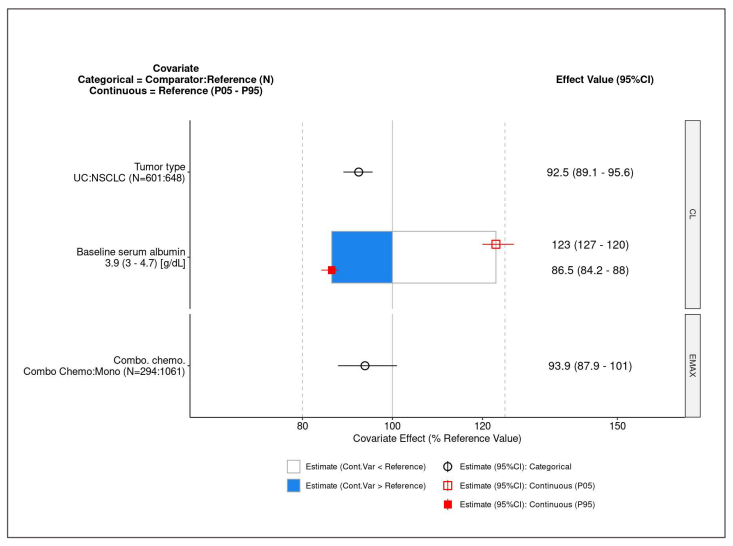
Covariate
Categorical = Comparator:Reference (N)
Continuous = Reference (P05 - P95)
Effect Value (95%CI)
Tumor type
UC:NSCLC (N=601:648)
92.5 (89.1 - 95.6)
Baseline serum albumin
3.9 (3 - 4.7) [g/dL]
123 (127 - 120)
86.5 (84.2 - 88)
Combo. chemo.
Combo Chemo:Mono (N=294:1061)
93.9 (87.9 - 101)
CL
EMAX
80
100
120
150
Covariate Effect (% Reference Value)
Estimate (Cont.Var < Reference)
Estimate (Cont.Var > Reference)
Estimate (95%CI): Categorical
Estimate (95%CI): Continuous (P05)
Estimate (95%CI): Continuous (P95)

Conclusions

- This work provides a comprehensive framework for characterization of the PK of a drug in new therapeutic settings, using nivolumab as a case study.
- Initially, prioritizing external evaluation methods is recommended to avoid unnecessary redundant model development, a particularly valuable approach when faced with tight deadlines.
- Model refinement is then undertaken only in the presence of significant bias, indicating that potential other factors may affect PK variability between studies and patient populations.

References

[1] OPDIVO® Package Insert. http://packageinserts.bms.com/pi/pi_opdivo.pdf
[2] OPDIVO® Annex I Summary of Product Characteristics. https://www.ema.europa.eu/en/documents/productinformation/opdivo-epar-product-information_en.pdf
[3] Zhang J, et al. Population Pharmacokinetics of Nivolumab in Combination With Ipilimumab in Patients With Advanced Malignancies. CPT Pharmacometrics Syst Pharmacol 2019 Dec;8(12):962-970.

Acknowledgments and Declaration of interests

This study was supported by Bristol Myers Squibb.

Presented at the 32nd Annual Meeting of the Population Approach Group in Europe (PAGE); 26-28 June 2024; Rome, Italy

Email: melanie@pumas.ai

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.