Preliminary Pharmacokinetic/Pharmacodynamic (PK/PD) Analysis for the Effect of Tanezumab on Overall Daily Pain Score Data in Adults with Moderate-to-Severe Pain due to Osteoarthritis of the Knee

Rujia Xie,[1] Rosalin Arends,[2] Stephen Olson,[2] Scott Marshall[1]

[1]Pfizer, Sandwich, UK; [2]Pfizer, New London, CT, USA

BACKGROUND

- Tanezumab is a humanized monoclonal IgG_2 antibody that binds to human nerve growth factor (NGF) with high affinity and specificity.
- Tanezumab is currently in development for the treatment of osteoarthritis, chronic low back pain, and other chronic pain conditions.

OBJECTIVE

- To characterize the exposure response (overall daily pain score [DPS]) relationship vs. time for tanezumab.

METHODS

Study Design

- Four hundred and forty-four osteoarthritis (OA) patients were treated in one of the following dose groups: placebo or 10, 25, 50, 100, or 200 μg/kg tanezumab administered as a 10-minute intravenous (IV) infusion on 2 occasions, 55 days apart (Day 1 and Day 56).
- Blood samples for the determination of tanezumab plasma concentrations were collected before and 1 and 2 hours after both doses of tanezumab (Days 1 and 56) as well as on Days 14, 28, 70, 84, 112, 136, and 182.
- DPS was recorded every evening on a visual analog scale (VAS) describing average pain over the past 24 hours from 0 (no pain) to 100 (high pain).

Modeling Strategy

- During the pharmacokinetic (PK)/pharmacodynamic (PD) model building, the individual PK parameter estimates were fixed.
- The PK/PD model used to describe the mean DPS profile over time was developed in stages—(1) the placebo model and (2) the base PK/PD model. The naïve pooled approach was employed.
- A Visual Predictive Check (VPC) was performed for the final PK/PD model to qualify the model with respect to the prediction of the mean DPS response. The original study was simulated 500 times.
- The fixed effect PD parameter estimates and associated uncertainty (covariance matrix of estimate) from the final model were used for the simulations. The PK/PD model developed for the average weekly pain score (WPS) using DPS data was then used for simulations to explore dose strategy and dose regimen.
- The PD model used was an indirect response model based on the assumption that tanezumab inhibits production of a pain stimulus, for example, that produced by NGF as measured by DPS. The PK/PD models are represented in **Figure 1**.

FIGURE 1: Schematic of a 2-compartment PK model and indirect response PD model

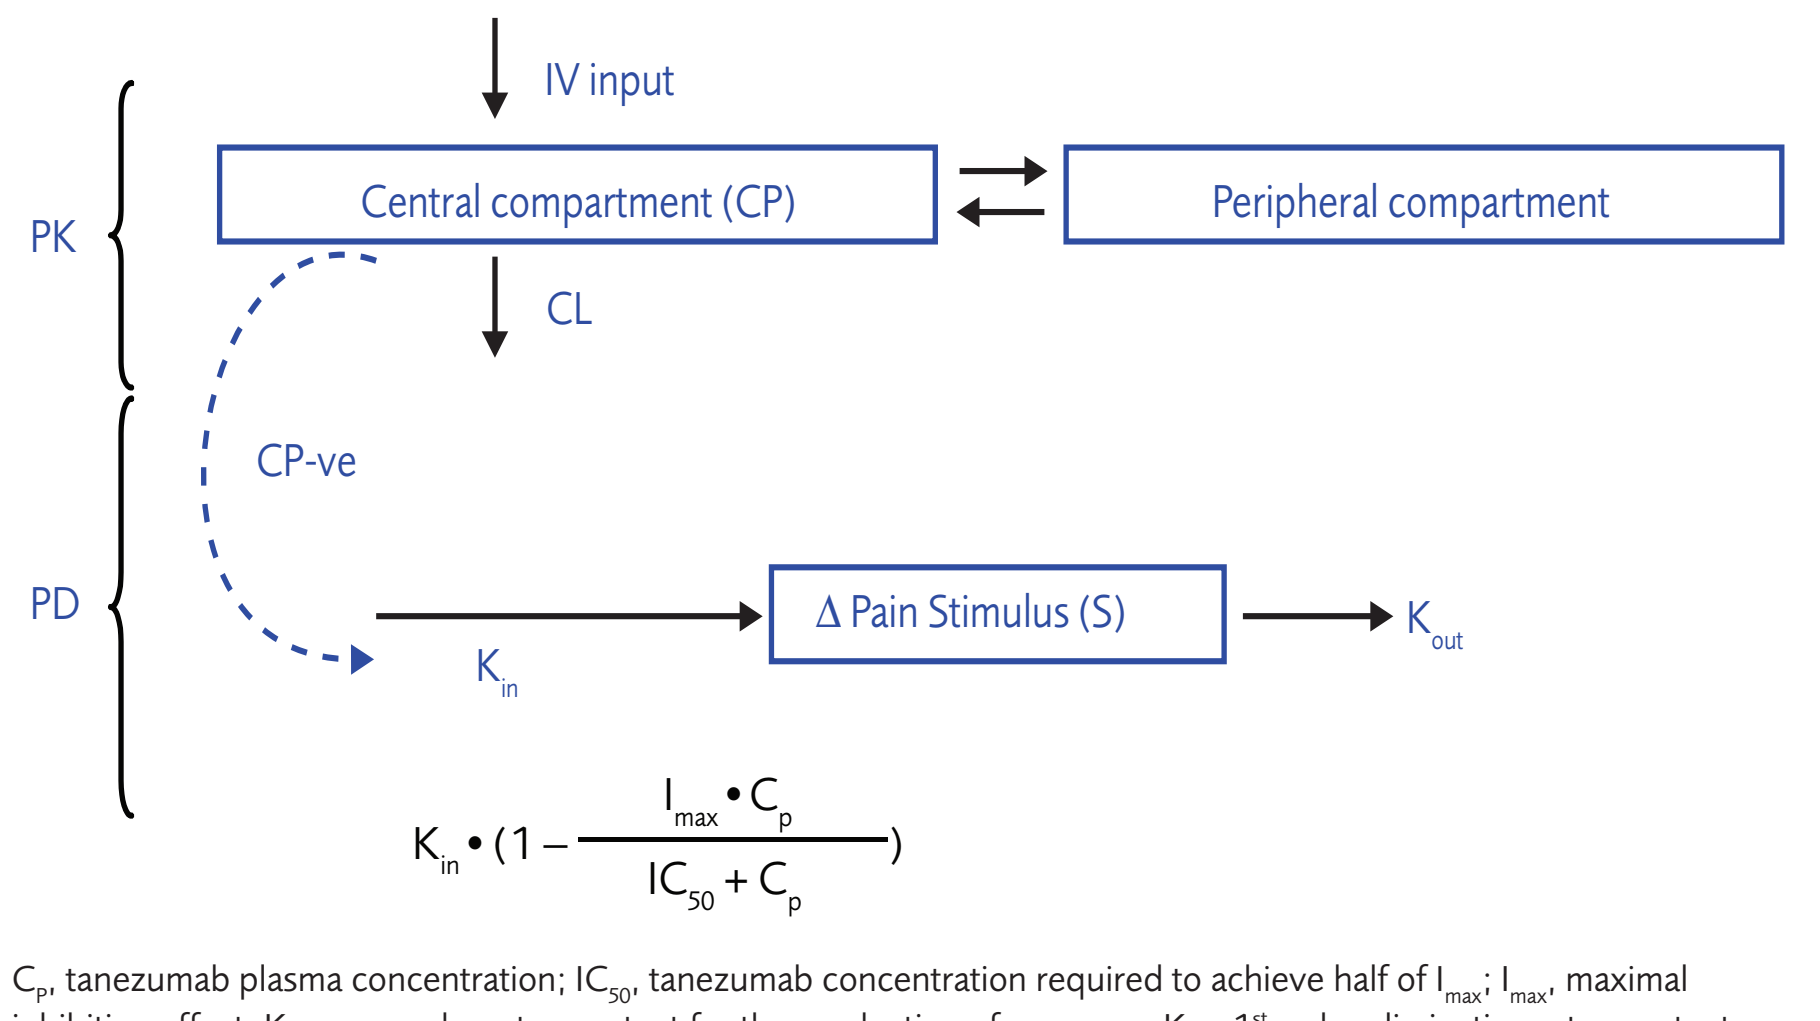
IV input
Central compartment (CP)
Peripheral compartment
CL
PK
CP-ve
PD
Δ Pain Stimulus (S)
K_{in}
K_{out}
$K_{in} \bullet (1 - \frac{I_{max} \bullet C_p}{IC_{50} + C_p})$

C_p, tanezumab plasma concentration; IC_{50}, tanezumab concentration required to achieve half of I_{max}; I_{max}, maximal inhibiting effect; K_{in}, zero-order rate constant for the production of response; K_{out}, 1st order elimination rate constant

- The PK/PD model assumed that the placebo and drug effects were proportional to baseline DPS:

$DPS = BASE \bullet (1\text{-}fp) \bullet fd$

where fp is placebo effect as a function of time; fd is drug effect as a function of tanezumab concentration; and BASE is baseline DPS.

- The transient dose-related attenuation of DPS that was observed approximately 14 days following the first dose of tanezumab (**Figure 2**) was described by a modified gamma distribution function as follows:

$$I_{max} = I_{max} \bullet \left(1 - scale \bullet \frac{t^{K_{off} \bullet T_{max}} \bullet e^{-K_{off} \bullet t}}{T_{max}^{K_{off} \bullet T_{max}} \bullet e^{-K_{off} \bullet T_{max}}} \bullet \frac{Dose}{Dose + ED_{50}}\right)$$

where ED_{50} is the dose required to reach half of maximal inhibition; K_{off} is the 1st order rate constant determining the shape of attenuation; I_{max} is maximal inhibiting effect; t is time; T_{max} is time to reach maximal attenuation; and scale is the scaling factor accounting for the proportional reduction of I_{max}.

- It was assumed that the reduction of I_{max} is related to dose as described by a maximum effect (E_{max}) model.

FIGURE 2: Mean overall change from baseline in DPS over time by treatment group

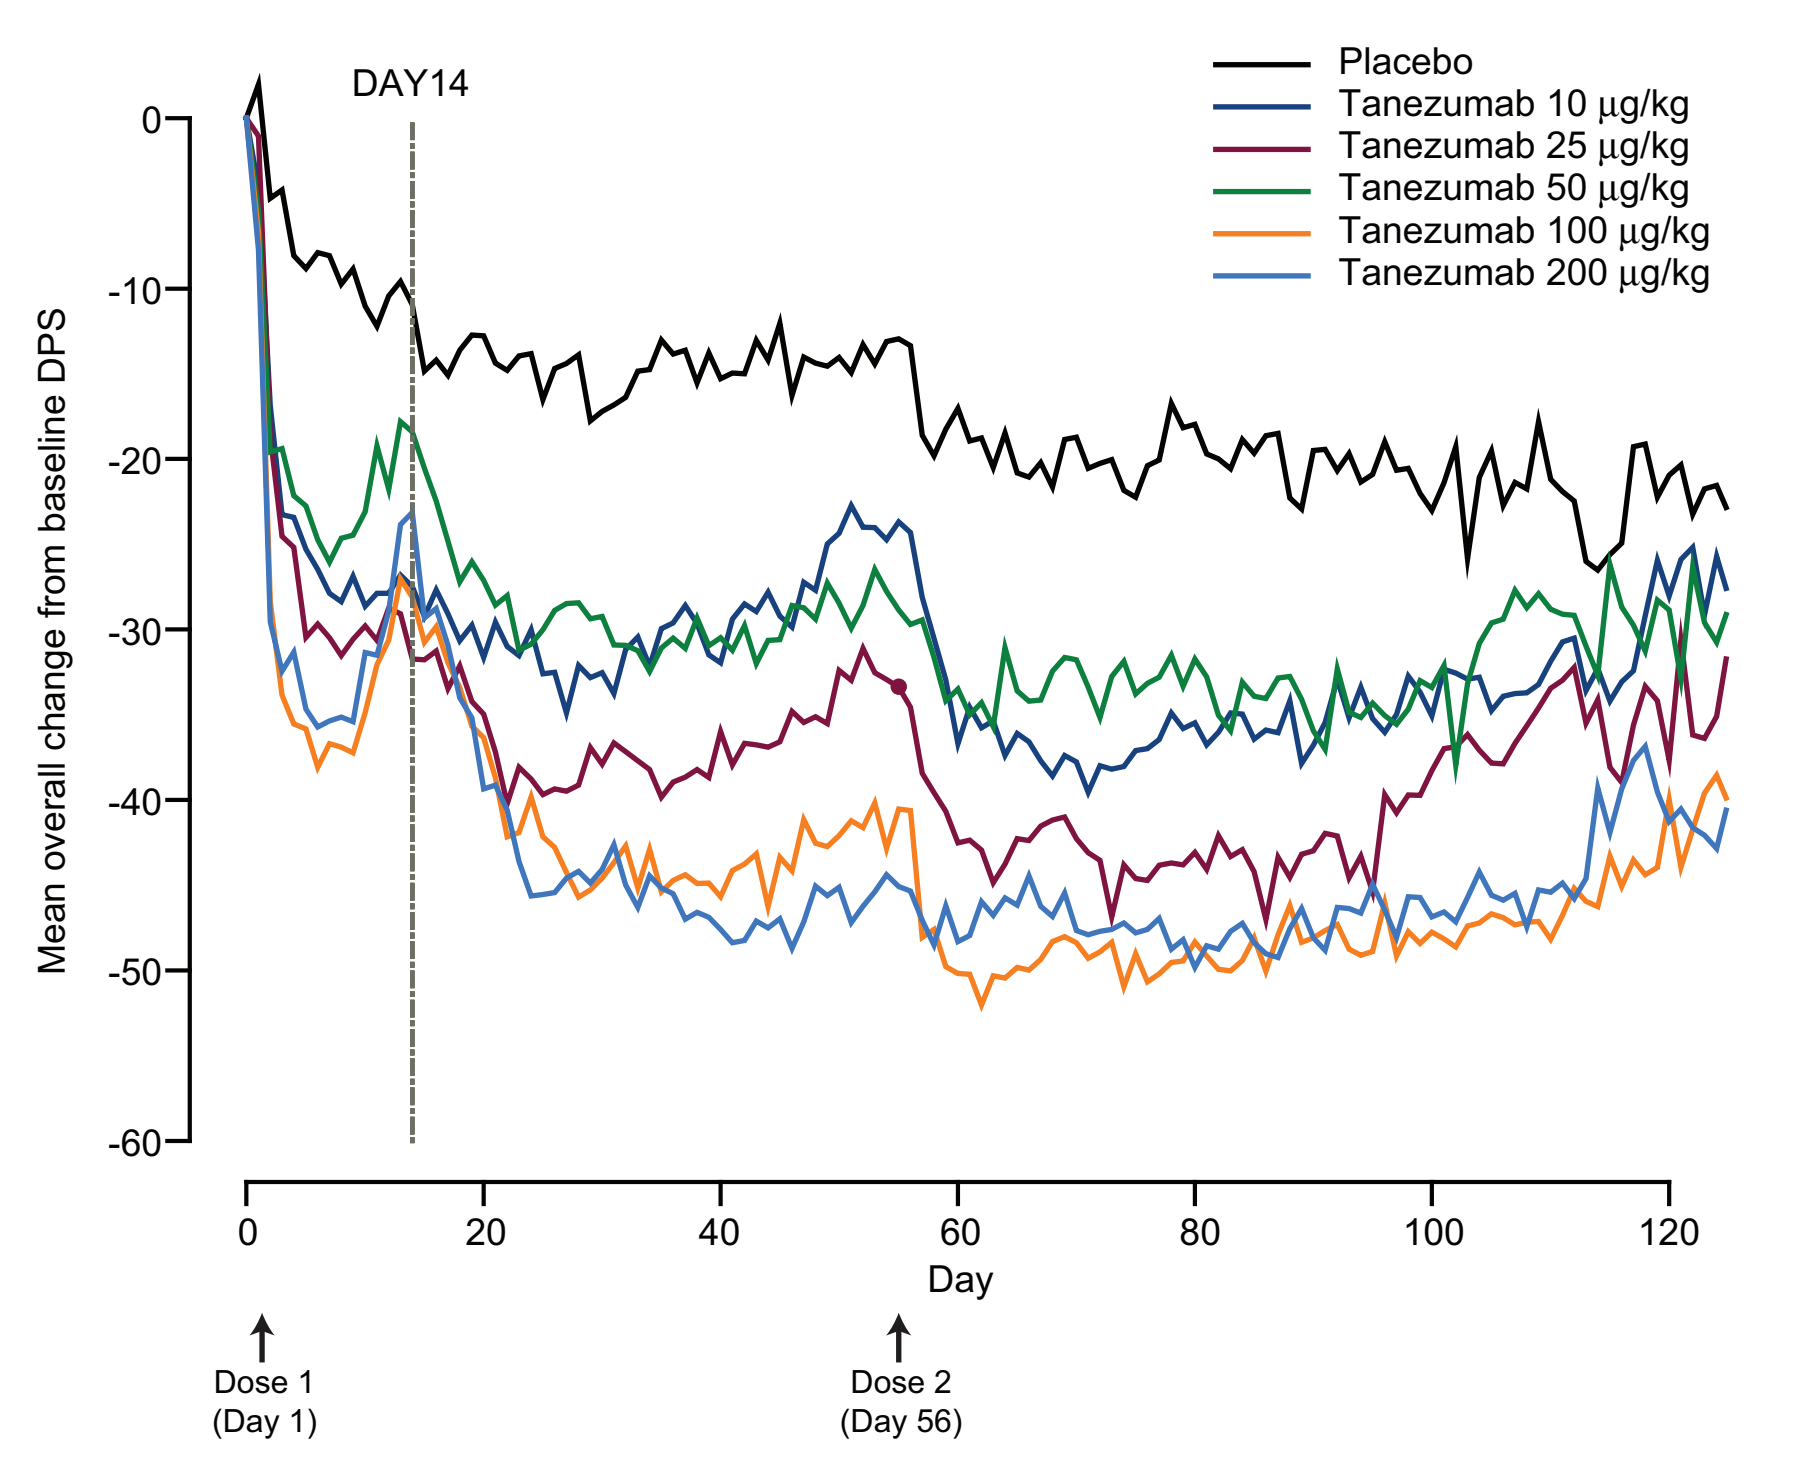
DAY14
Placebo
Tanezumab 10 μg/kg
Tanezumab 25 μg/kg
Tanezumab 50 μg/kg
Tanezumab 100 μg/kg
Tanezumab 200 μg/kg
Mean overall change from baseline DPS
Day
Dose 1 (Day 1)
Dose 2 (Day 56)

RESULTS

- The PK of tanezumab was well described by a 2-compartment model.
- Body weight was found to be a significant predictor of clearance and volume but only explained 4% of the overall 47% unexplained inter-subject variability.[1]
- Placebo effect was well described by an exponential time-dependent model. The onset of placebo effect was relatively fast with an equilibrium half-life of 7.7 days after the first dose. The maximum placebo effect was estimated to be 25.2% of the baseline value.
- An indirect response model was found to best characterize the relationship between tanezumab concentration and response (**Figure 3**), and the observed attenuation of DPS was captured by this model.
- The drug effect was characterized by an inhibitory E_{max} model, which was expressed in terms of I_{max}, and the tanezumab concentration required to achieve half of I_{max} (IC_{50}). I_{max} and IC_{50} were estimated to be 0.538 and 69.3 ng/ml, respectively. The major PD parameter estimates are presented in **Table 1**.

FIGURE 3: Mean predicted (red line) and observed (black dots) DPS by day for each treatment group

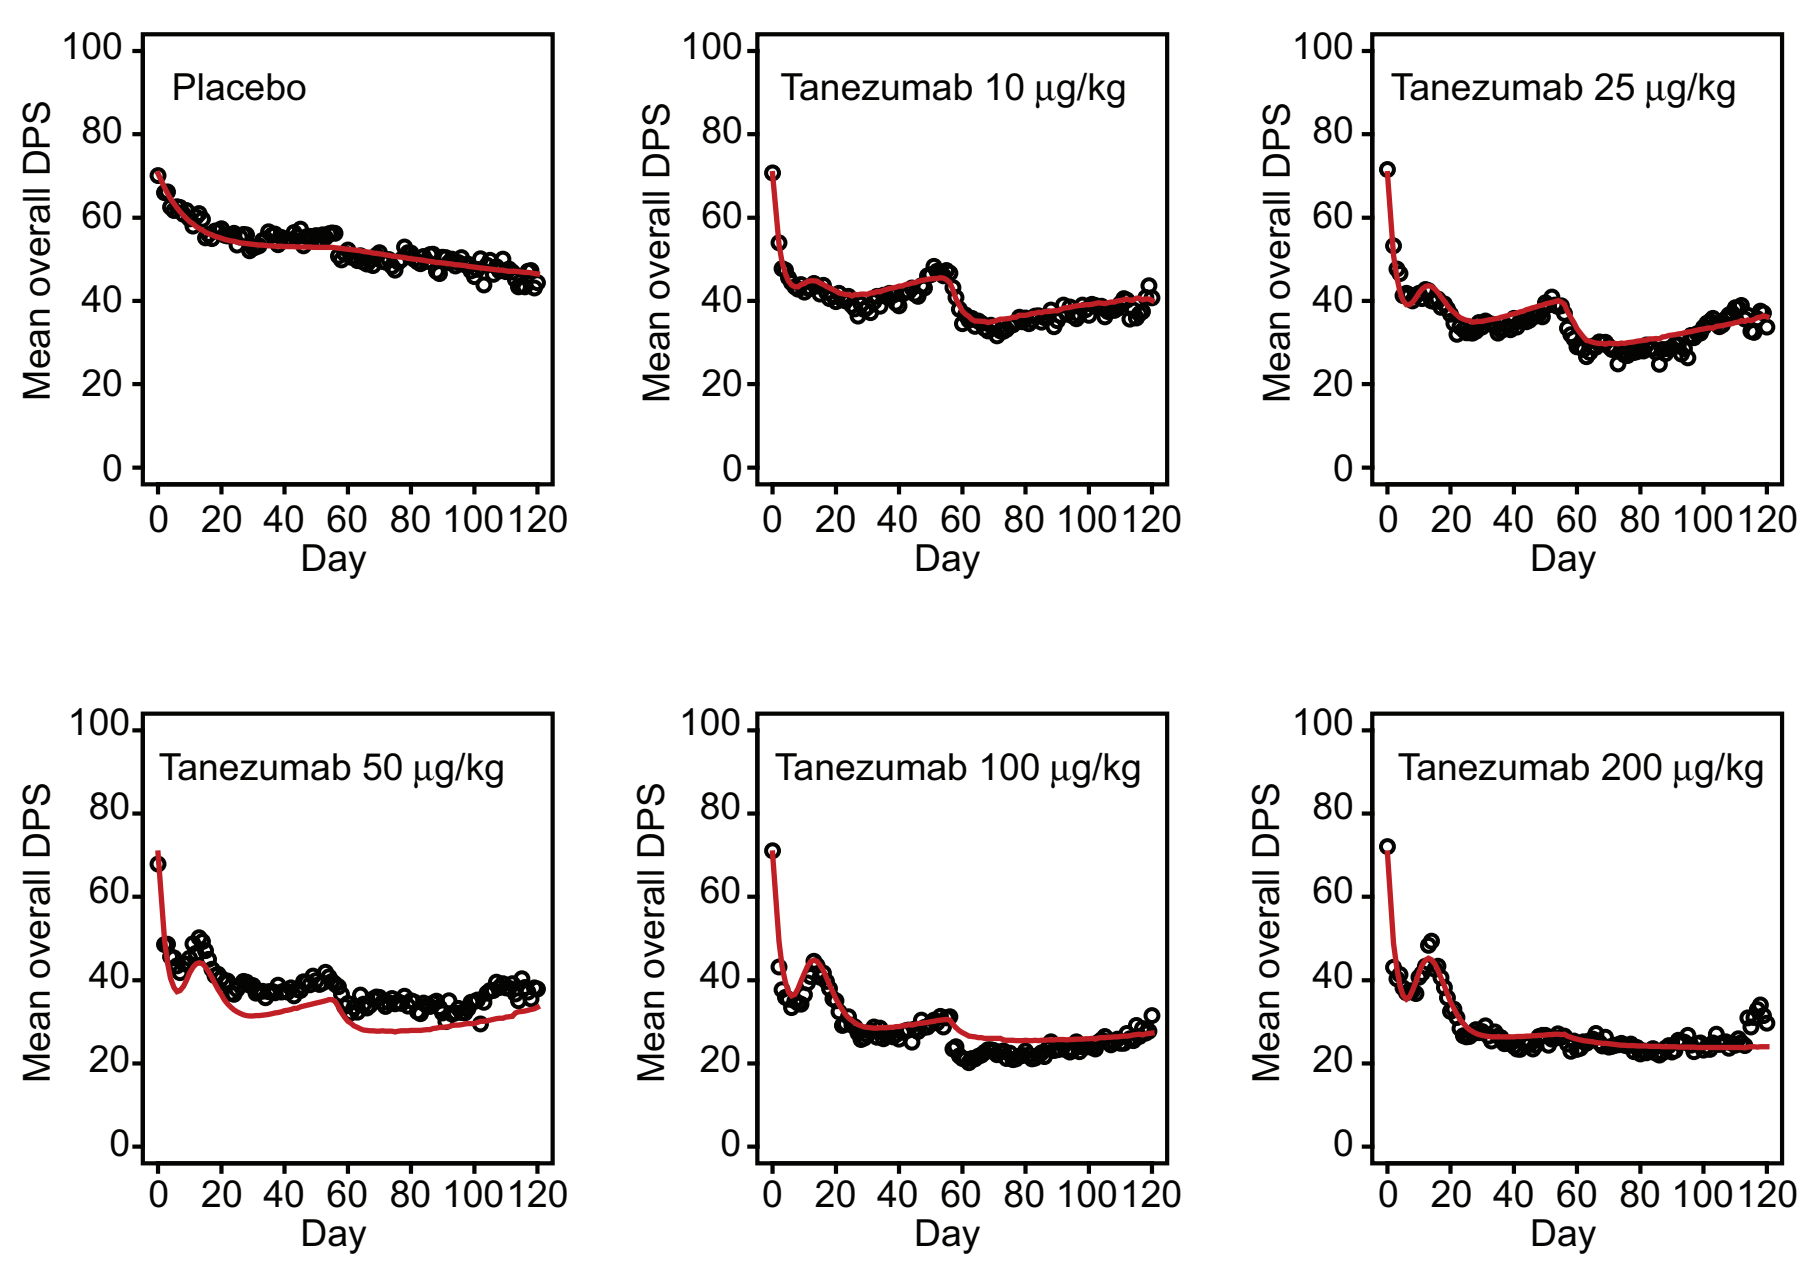
Placebo
Tanezumab 10 μg/kg
Tanezumab 25 μg/kg
Tanezumab 50 μg/kg
Tanezumab 100 μg/kg
Tanezumab 200 μg/kg
Mean overall DPS
Day

TABLE 1: PD parameter estimates (with relative standard error [RSE]%)

Parameter	Parameter estimates (RSE%)
BASE	70.5 (0.8)
P_{max}	0.252 (16.1)
K_f (day^{-1})	0.103 (23)
I_{max}	0.538 (9.4)
IC_{50} (ng/ml)	69.3 (48.5)
K_{out} (day^{-1})	0.366 (16.2)
T_{max} (day)	11.6 (4.3)
K_{off} (day^{-1})	0.712 (29.4)
ED_{50} (μg/kg)	11.8 (77.2)
Scale	0.729 (17.1)
Residual	24.6 (2.0)

BASE, baseline DPS; ED_{50}, the dose required to reach half of maximal inhibition; IC_{50}, tanezumab concentration required to achieve half of I_{max}; I_{max}, maximal inhibiting effect; K_f, 1st order rate constant; K_{off}, rate constant of maximal attenuation; K_{out}, 1st order elimination rate constant; P_{max}, maximum placebo effect; scale, the scaling factor accounting for the proportional reduction of I_{max}; T_{max}, time to reach maximal attenuation

- The plots of VPC indicate that the observed mean DPS (with 90% confidence intervals [CI]) were well predicted by the model (**Figure 4**).
- The predicted mean change from baseline in WPS dose response, showing the largest and smallest effects for dosing intervals of 6, 8, and 12 weeks, demonstrates that efficacy is maintained across an 8-week dosing interval (**Figure 5**).

FIGURE 4: Visual predictive check of mean DPS from the final model (n = 500); observed mean (blue line) with 90% confidence intervals (CI) (orange lines) and simulated mean (black line) with 90% predicted intervals (PI) (green band)

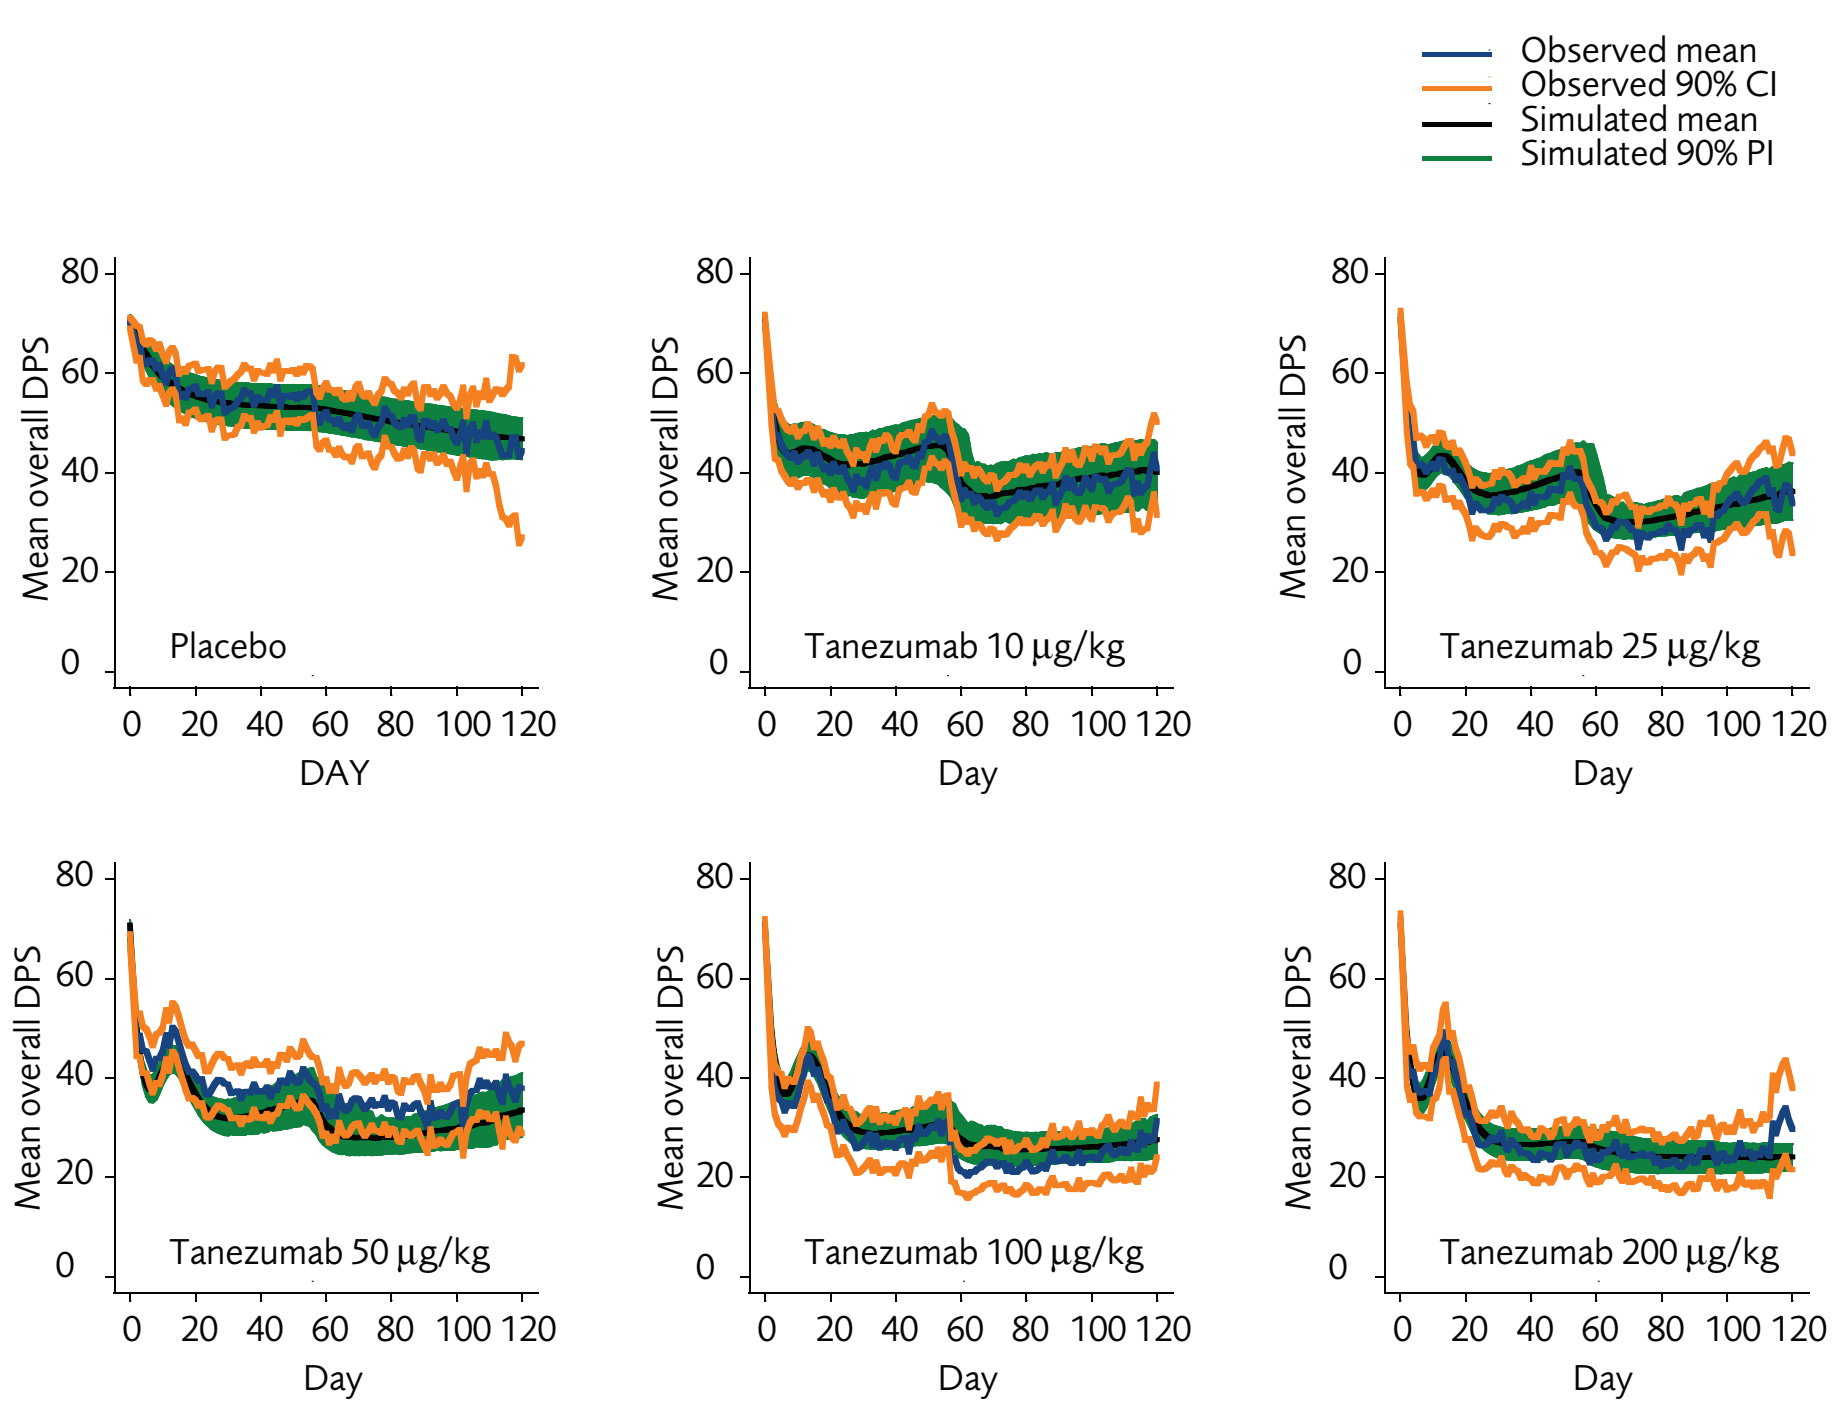
Observed mean
Observed 90% CI
Simulated mean
Simulated 90% PI
Placebo
Tanezumab 10 μg/kg
Tanezumab 25 μg/kg
Tanezumab 50 μg/kg
Tanezumab 100 μg/kg
Tanezumab 200 μg/kg
Mean overall DPS
DAY
Day

FIGURE 5: Predicted mean change from baseline in WPS dose response (n = 1000) showing the largest (peak) and smallest (trough) effects during the 4th dose interval for 6, 8, and 12-weekly administration of tanezumab

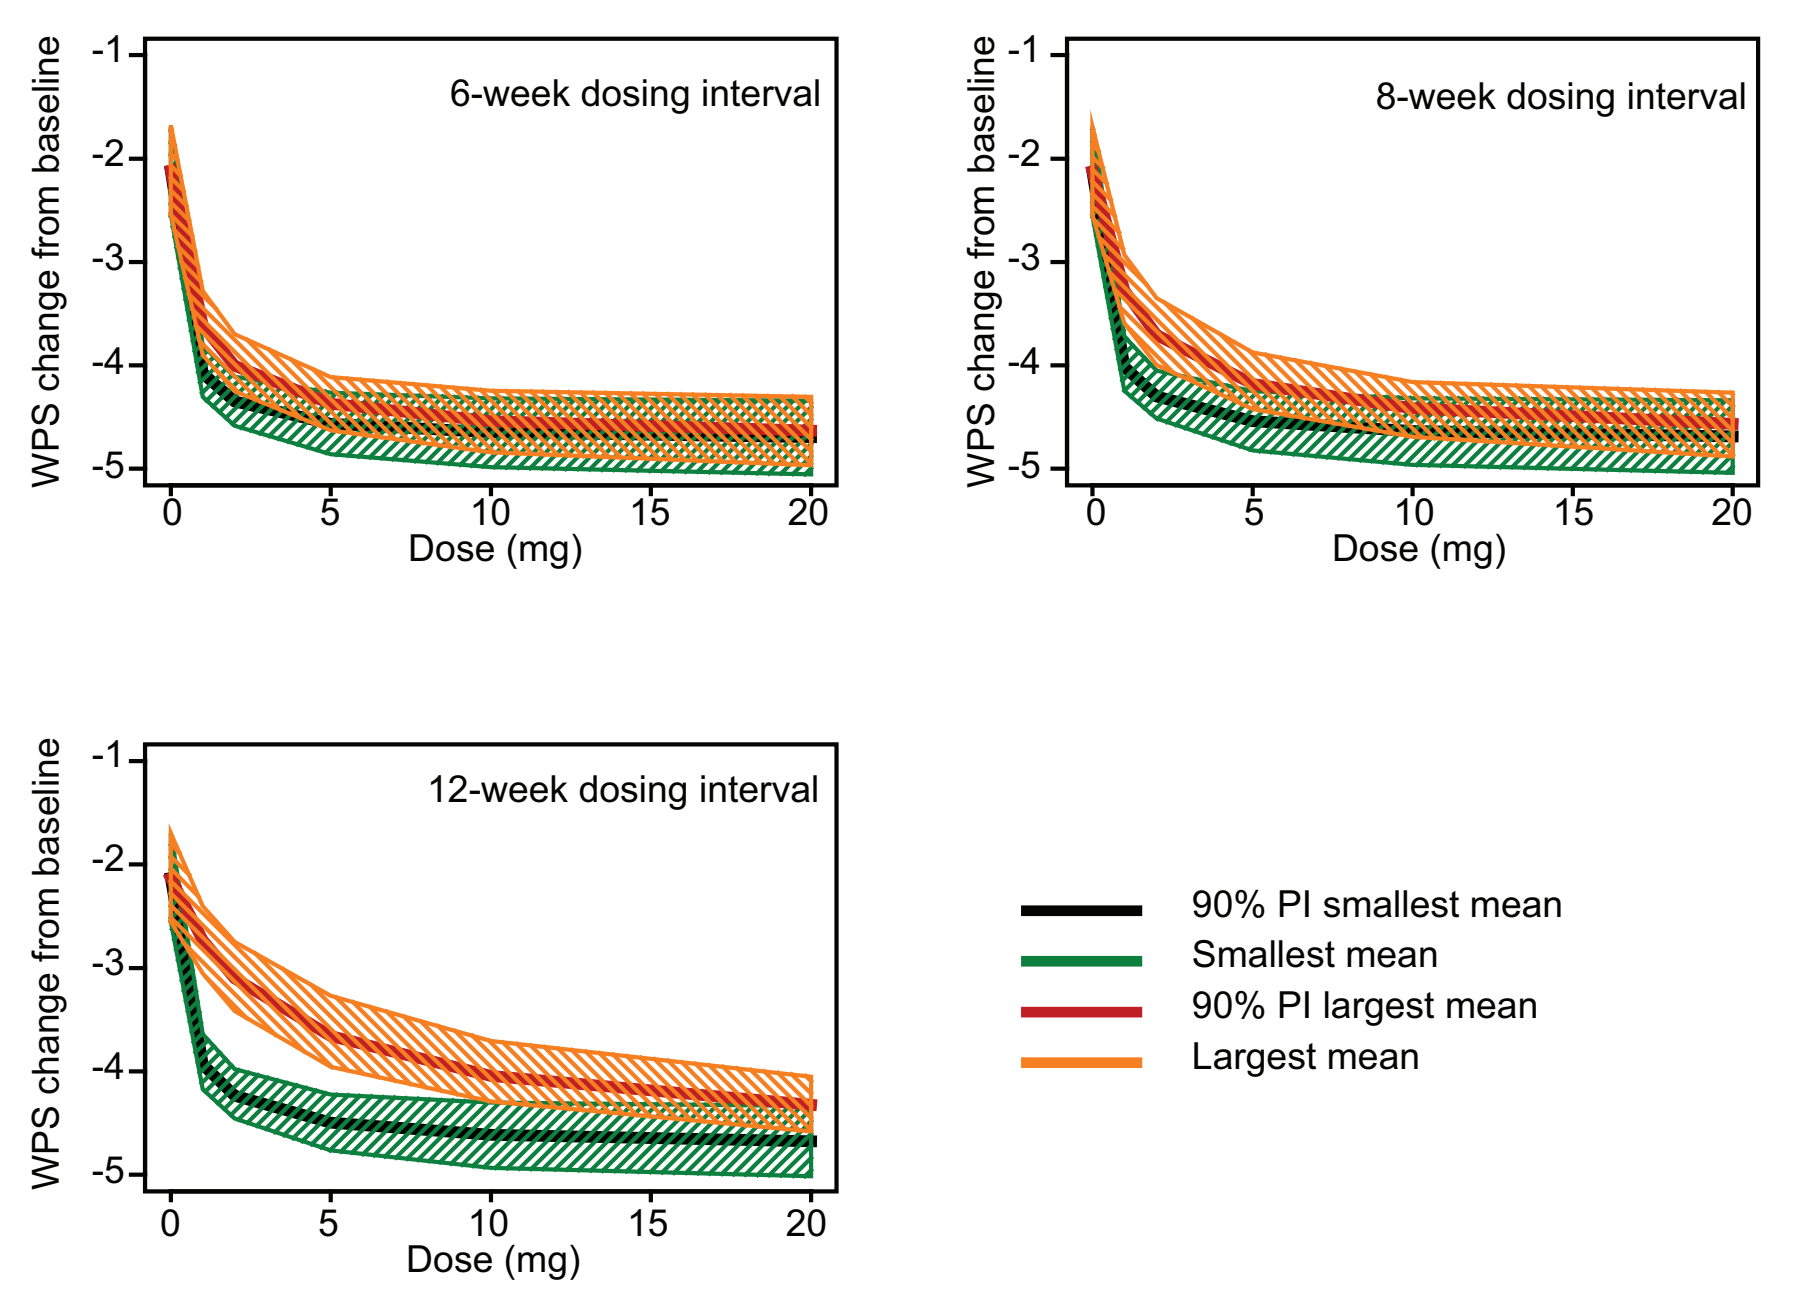
6-week dosing interval
8-week dosing interval
12-week dosing interval
WPS change from baseline
Dose (mg)
90% PI smallest mean
Smallest mean
90% PI largest mean
Largest mean

DISCUSSION

- The model used here was limited by the assumption that each DPS observation was independent, which was not the case. The model can be improved by:
 - Incorporating an auto-correlation.
 - Investigating the random effects model and improving the skewness of inter-individual variability.
 - Incorporating a mechanistic component of the biology of NGF to describe the observed attenuation of DPS.
 - Extending work to other clinical pain states.

CONCLUSIONS

- The DPS data were adequately described by the proposed semi-mechanistic PK/PD model.
- Subsequent simulations using the PK/PD model support the use of an 8-week dosing regimen with fixed doses of 2.5, 5, and 10 mg tanezumab in Phase 3 studies in OA patients.

REFERENCES 1. Arends R, Lalovich B, Marshall S, et al. Population PK modeling to support the use of a fixed dosing regimen in phase 3 for tanezumab, an anti-NGF humanized antibody. *AAPS NBC*, 2009.

ACKNOWLEDGEMENTS We would like to thank Barry Weatherley for his valuable contributions to the modeling work.

DISCLOSURE This study was supported by Pfizer Inc. Editorial support for the production of this poster was provided by Dr Aideen Young of UBC Scientific Solutions and funded by Pfizer Inc.

Poster presented at the 2009 annual meeting of the Population Approach Group in Europe (PAGE); June 23–26, 2009; St Petersburg, Russia