A pharmacokinetic-pharmacodynamic model for ECG pattern changes in dog and monkey

F. Jonsson(1), A. Fleury(2), T. Lavé(2)

(1) Pharsight, a Certara company, St. Louis, MO, USA;
(2) F. Hoffmann-La Roche Ltd., Modeling and Simulation Group, Drug Metabolism and Pharmacokinetics Department, Basel, Switzerland

1 - Introduction

R1551 is a dual NK1-NK3 antagonist under development for the treatment of schizophrenia. During safety assessments of R1551, delays in AV conduction and ventricular depolarisation were observed in two non-rodent species. Telemetry studies were conducted in dogs, and monkeys to identify any species-specific effect. In dogs, statistically significant PR-interval prolongation and QRS-complex widening were observed, starting at 10 mg/kg. Isolated sporadic second-degree Wenckebach AV blocks were seen in two male dogs at 30 mg/kg, and in all male dogs at 100 mg/kg. In monkeys, R1551 also prolonged PR and QRS at 30 mg/kg, but no second-degree or higher AV block was observed. Neither assays of receptor or ion channel binding nor functional ion channel tests (assessing hERG current in transfected CHO cells, and fast sodium and L-type calcium currents in isolated human cardiomyocytes) with the parent compound could explain the in vivo findings.

In addition to these ECG changes in relevant species and their lack of a mechanistic explanation, we observed high inter-individual variability in exposure, and no clear relationship between plasma drug concentrations and ECG effects. We therefore decided to assess the relationship between R1551 exposure and prolongation of PR and QRS intervals using a pharmacokinetic–pharmacodynamic approach, and to elucidate possible causes for such changes.

Objectives:
To determine the concentration–time profile of R1551 and its effect on the ECG using non-linear mixed-effects modeling, and to evaluate the usefulness of this approach.

2 - Methods

Pharmacokinetic-pharmacodynamic data:

• Beagle Dog Pharmacokinetics
1. 1 mg/kg single IV (n=4)
2. 30 mg/kg single PO (n=4)
3. *0, 3, 10, 30 and 100 mg/kg crossover PO (n=8)*
4. 0, 5, 15, 30 or 45 mg/kg daily PO over six weeks (n=22)
5. 0, 5, 25 or 75 mg/kg daily PO over two weeks (n=6)

• Cynomolgus Monkey Pharmacokinetics
6. 0.75 mg/kg single IV (n=2)
7. 10 mg/kg single PO (n=8)
8. *0, 10, 30 and 100 mg/kg crossover daily PO (n=7), each dose given 5 days*

• Pharmacodynamics (studies 3 and 8)
Telemetry used to measure ECG parameters (PR interval and QRS complex) and heart rate up to ≥ 24 h after the final dose.

Method:
Use NONMEM to develop population-based model for the pharmacokinetics, based on available data in each species
Use NONMEM-predicted PK profiles (concentrations) as predictors (time-varying covariates) of the PR and QRS profiles in dog and monkey when developing models for the telemetry data in a similar fashion as for pharmacokinetics

3 - Results: model parameters for PK in dog and monkey

PK in Dog:
2-compartment model w/ first-order absorption worked well
Interoccasion variability on bioavailability (F) essential

Parameter	Estimate	SD	Population variability (%CV, lognormal)	Interoccasion variability (%CV, lognormal)
Clearance (l/kg/h)	0.370	0.0346	9.8	
Vc (l/kg)	0.754	0.221	21	
ka (h-1)	0.236	0.0629		
F	0.419	0.0756		29
Q (l/kg/h)	0.306	0.0585		
Vp (d-1)	8.58	1.17		
ε (proportional residual error)	0.106	0.0198		

Predicted R1551 concentrations in a representative dog:

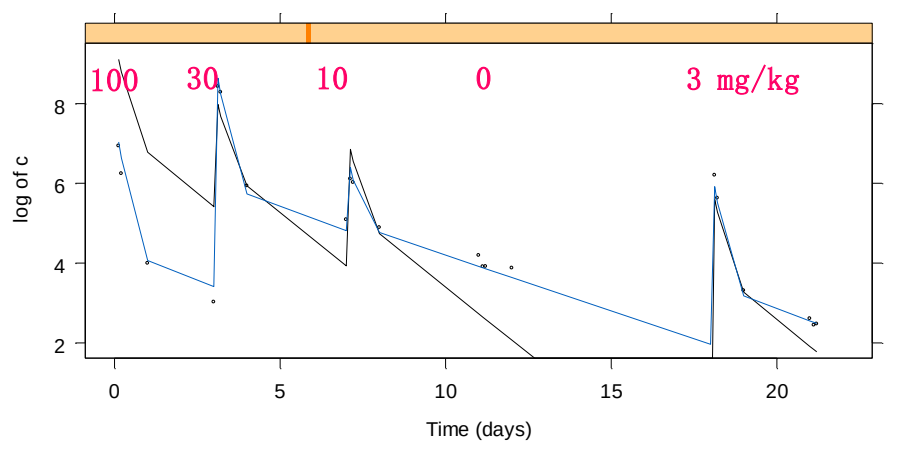
100
30
10
0
3 mg/kg
log of c
Time (days)

PK in Monkey:
2-compartment model w/ first-order absorption worked well
Nonlinear kinetics with variability in both km and Vmax essential

Parameter	Estimate	SD	Population variability (%CV, lognormal)
Vmax (l/kg/h)	340	114	46
Km	448	131	34
Vc (l/kg)	1.37	0.418	108
ka (h-1)	0.125	0.00600	
F	-1.47	0.375	86
Q (l/kg/h)	0.0128	0.00774	
Vp (d-1)	1.67	1.01	
ε (proportional residual error)	0.195	0.0327	

Large SDs

Predicted R1551 concentrations in a representative monkey:

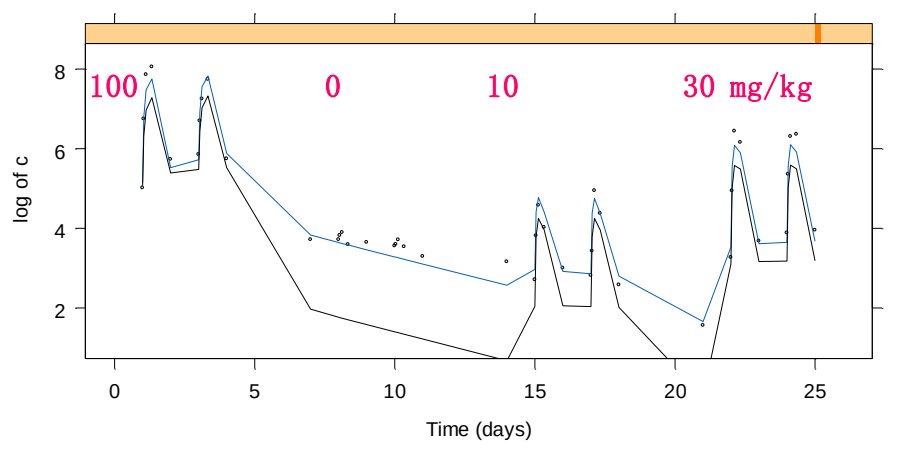
100
0
10
30 mg/kg
log of c
Time (days)

4 - Telemetry modelling

• Linear concentration-effect models provided a good fit to the observed data for both ECG parameters in dogs and QRS data in Monkey
 - In Monkey, the effect was small
 - The effect compartment was then unnecessary and removed

• Fit of PR data in Monkey improved by the addition of a placebo effect compartment, accounting for a suspected vehicle effect.

• Estimated slope factors:
 - PR interval – 0.0093 (dog) and 0.00934 ms mg-1 kg-1 l-1 (monkey)
 - QRS complex – 0.00274 (dog) and 0.00200 ms mg-1 kg-1 l-1 (monkey)

Conclusions

- The slope factors are similar between dog and monkey
 - Effect on the ECG pattern appears to be similar in magnitude between species
 - Time course of effect is different between species and endpoint
 - The similarity between species suggests that a similar magnitude of ECG change could be expected in humans given a similar metabolite pattern
- The PK/PD modeling approach explained the effect on the ECG pattern as a function of drug exposure

5 - Model parameters for PD in dog and monkey

Model parameters for PD in dog:

Parameter	Estimate	SD	Population variability (%CV, lognormal)
PR			
ke0	0.0892	0.0112	11
Baseline PR	95.1	2.74	0.64
slope	0.00930	0.00133	17
ε (proportional residual error)	0.00593	0.00137	
QRS			
ke0	0.122	0.0185	4.2
Baseline QRS	42.5	1.24	0.66
slope	0.00274	0.00101	250
ε (proportional residual error)	0.00742	0.00119	

PR profiles in a representative male dog:

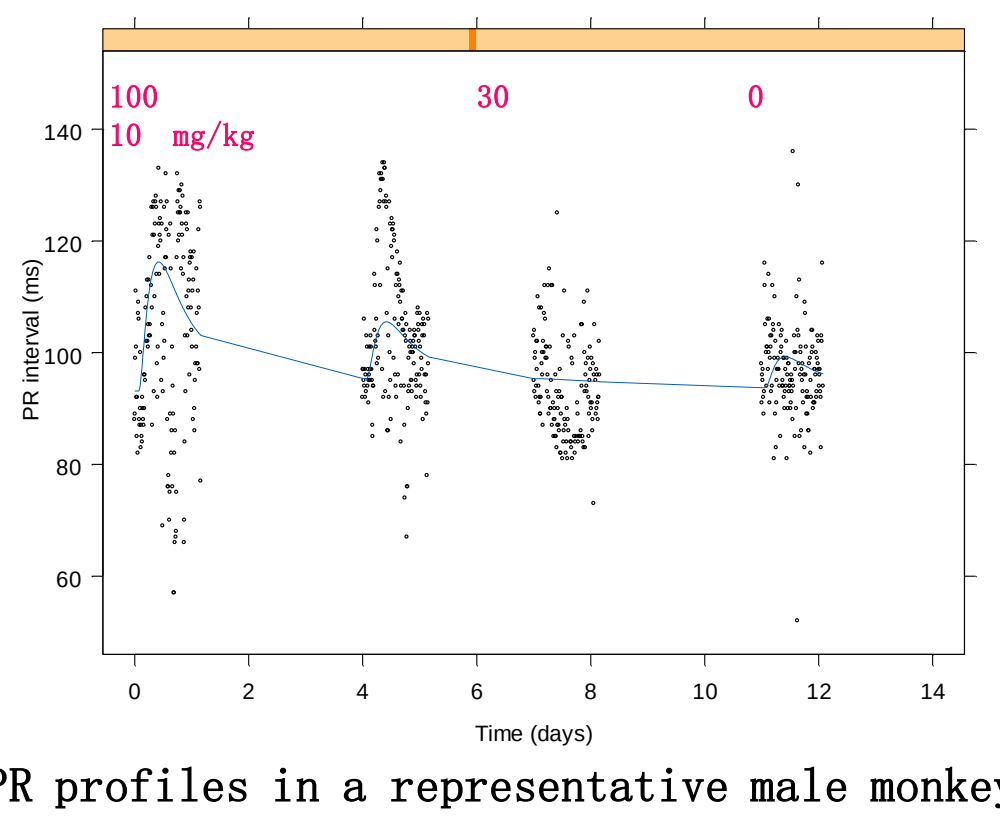
100
10 mg/kg
30
0
PR interval (ms)
Time (days)

QRS profiles in a representative male dog:

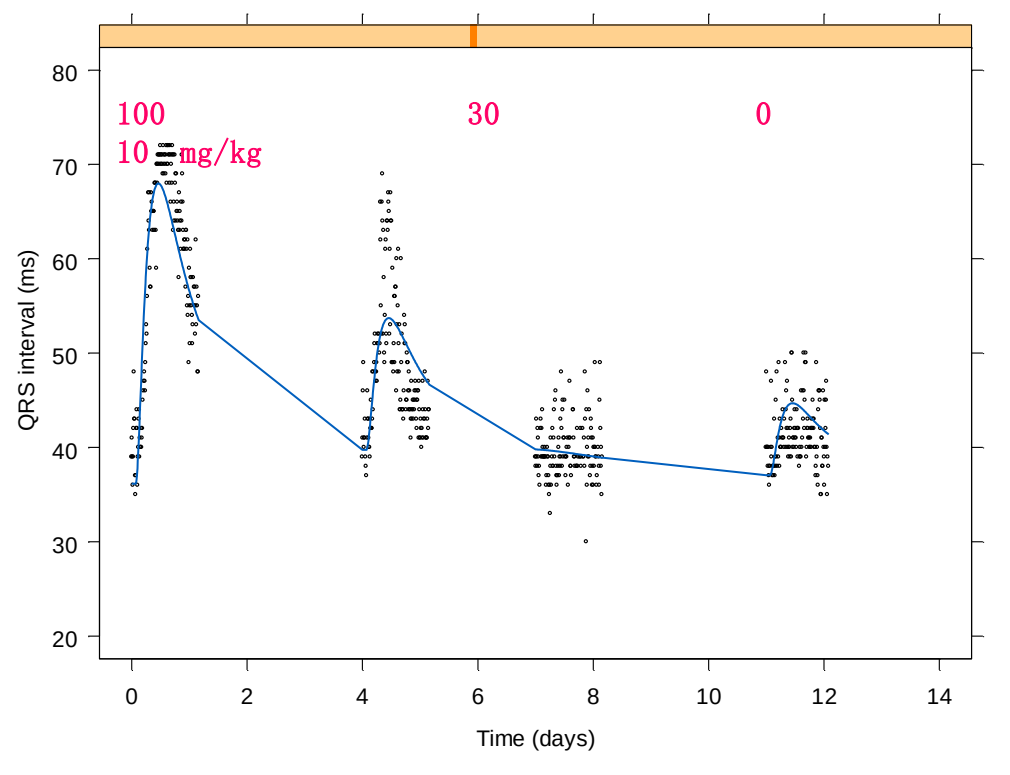
100
10 mg/kg
30
0
QRS interval (ms)
Time (days)

Model parameters for PD in monkey:

Parameter	Estimate	SD	Population variability (%CV, lognormal)
PR			
ke0	0.410	0.163	
Baseline PR	64.9	1.46	0.37
Slope	0.00934	0.00141	
ke0, placebo	0.111	0.00140	
slope, placebo	23.2	5.46	35
ε (proportional residual error)	0.00378	0.00025	
QRS			
Baseline QRS	46.5	1.11	0.4
slope	0.00200	0.000552	59
ε (proportional residual error)	0.00741	0.000066	

PR profiles in a representative male monkey:

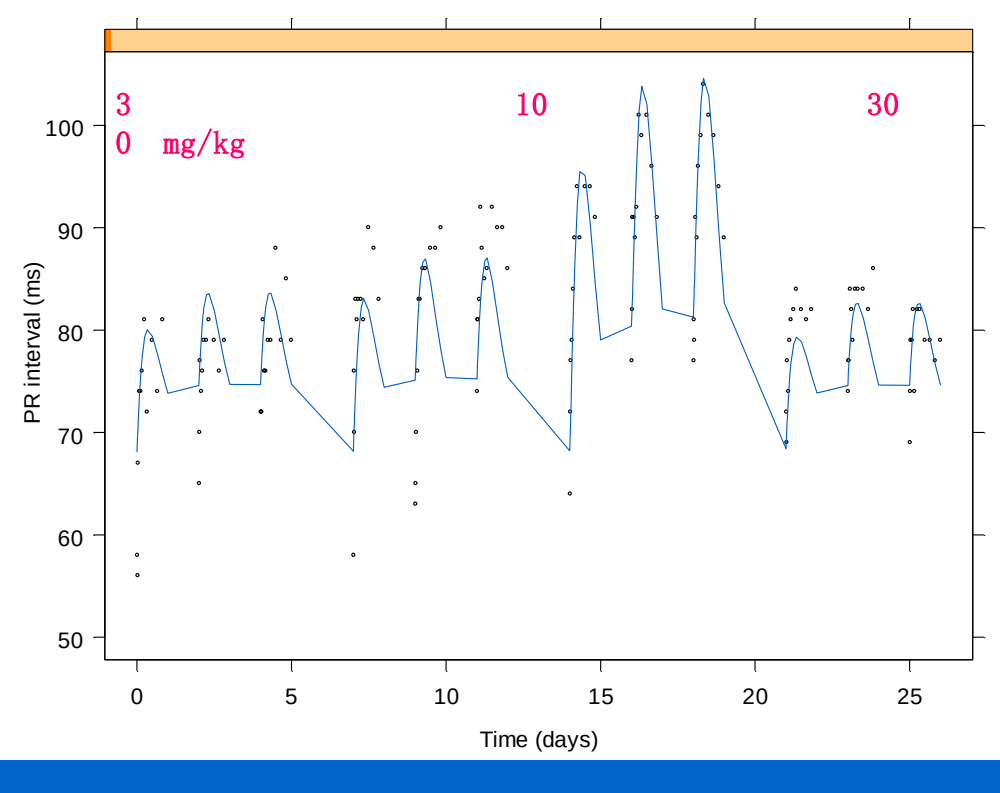
3
0 mg/kg
10
30
PR interval (ms)
Time (days)

QRS profiles in a representative male monkey:

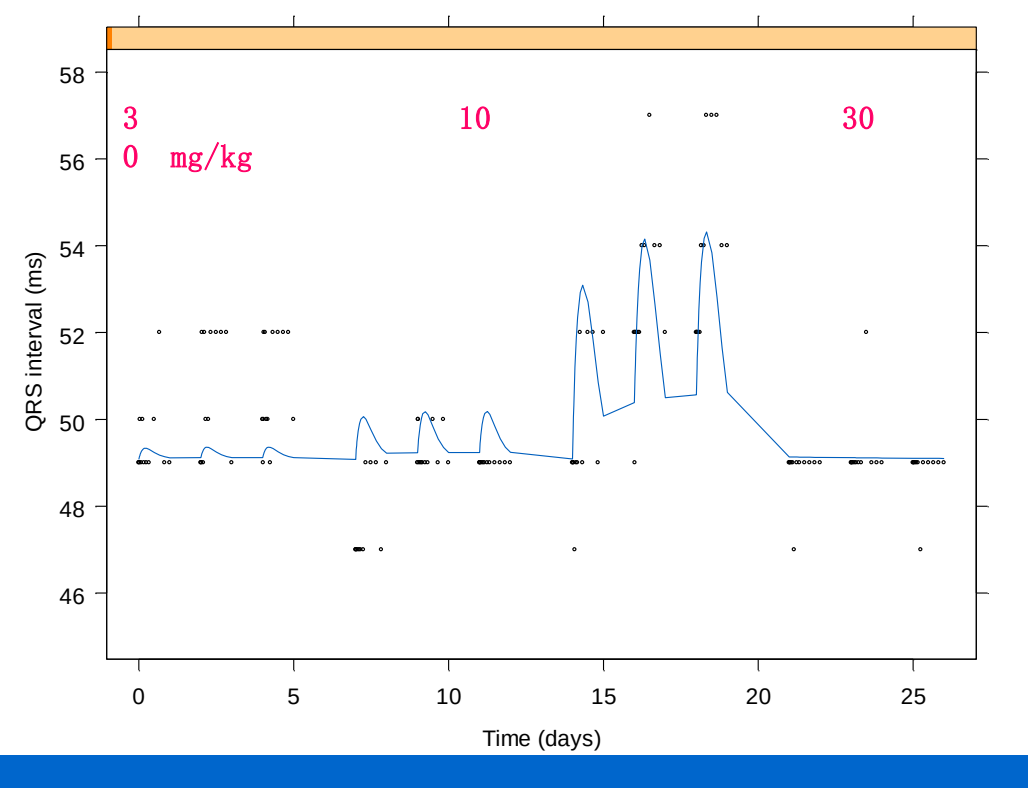
3
0 mg/kg
10
30
QRS interval (ms)
Time (days)