An assessment of the prediction accuracy of two IVIVC modelling methodologies

Clare Gaynor (1), Adrian Dunne (1) and John Davis (2)

1) UCD School of Mathematical Sciences, University College Dublin, Ireland
Scoil na nEolaíochtaí Matamaitice UCD, An Coláiste Ollscoile, Baile Átha Cliath, Éire
2) Clinical Pharmacology, Pfizer Global Research and Development, Sandwich, England

INTRODUCTION:

Information gathered during *in vitro* dissolution studies can be used, by means of a mathematical or statistical model, to predict the *in vivo* performance of a product e.g. plasma concentration of drug. This model has a number of applications including reducing the number of human studies necessary during drug development, setting specification limits for batch approval during routine manufacturing and acting as a substitute for the human studies required for regulatory approval. As a result, the accuracy and reliability of the predictions made by these *In Vitro– In Vivo* Correlation (IVIVC) models is of the utmost importance and substantial effort and resources go in to their development.

The accuracy with which *in vitro* observations can be used to predict an *in vivo* response may depend on the choice of method used to develop such a model. Two approaches to developing these models are predominantly used: a traditional deconvolution-based method and an alternative convolution-based technique. In spite of the fact that there are a number of areas of concern regarding the deconvolution-based methods[1], they are routinely used. The convolution-based alternative[2], which does not suffer from the same weaknesses is, however, relatively rarely employed. It has previously been shown[3] that, where an IVIVC relationship does exist, a deconvolution-based method is more likely than a convolution-based procedure to fail the FDA validation test[4] (32% and 0.1% failure rates respectively). The aim of this study is to supplement these results by further investigating the performance of both a deconvolution and a convolution based method and quantifying any difference in accuracy of prediction.

METHOD:

A simulation study to compare the conventional deconvolution based methods of establishing an IVIVC to an alternative non-linear mixed effects modelling approach was undertaken. In practise, the Extended Release (ER) dosage units of interest are dissolved *in vitro* and the fractions which have dissolved are recorded at a series of time points. ER dosage units from the same batch are then administered to a number of human subjects and their plasma drug concentrations are measured over time - these data contain information on dissolution, absorption, distribution, and elimination of the drug. A reference dose, which dissolves instantly, is administered to each of the same group of subjects and the resulting plasma drug concentrations are repeatedly measured for a predetermined period. These three kinds of data: *in vitro*, *in vivo* and reference, are used to establish the IVIVC model. The current project involves simulating such an IVIVC study for which the true model and parameter values are known.

In vitro and *in vivo* (extended release and reference) data were simulated for four formulations of a drug according to a model which incorporated an IVIVC relationship. The data were simulated as follows: let $F_{1i}(t)$ be the true fraction of drug dissolved from the i^{th} tablet of formulation h at time t *in vitro*, then the observed fraction dissolved is given by

$$Y_{h1i}(t) = \phi_1 F_{h1i}(t) + \varepsilon_{1i}(t) \qquad \varepsilon_{1i}(t) \sim N(0, \sigma_1^2)$$

with $\quad logit\,(F_{h1i}(t)) = logit\,(F_{h1}(t)) + u_i \qquad u_i \sim N(0, \omega_1^2)$

and $\quad F_{h1}(t) = 1\text{-}exp\,(-\lambda_{1h}t)$

where the tablet-to-tablet variation is given by ω_1^2, the intra-tablet variation by σ_1^2, λ_{1h} determines the rate of dissolution of the drug in formulation h and ϕ_1 accounts for any difference between true dose and label claim.

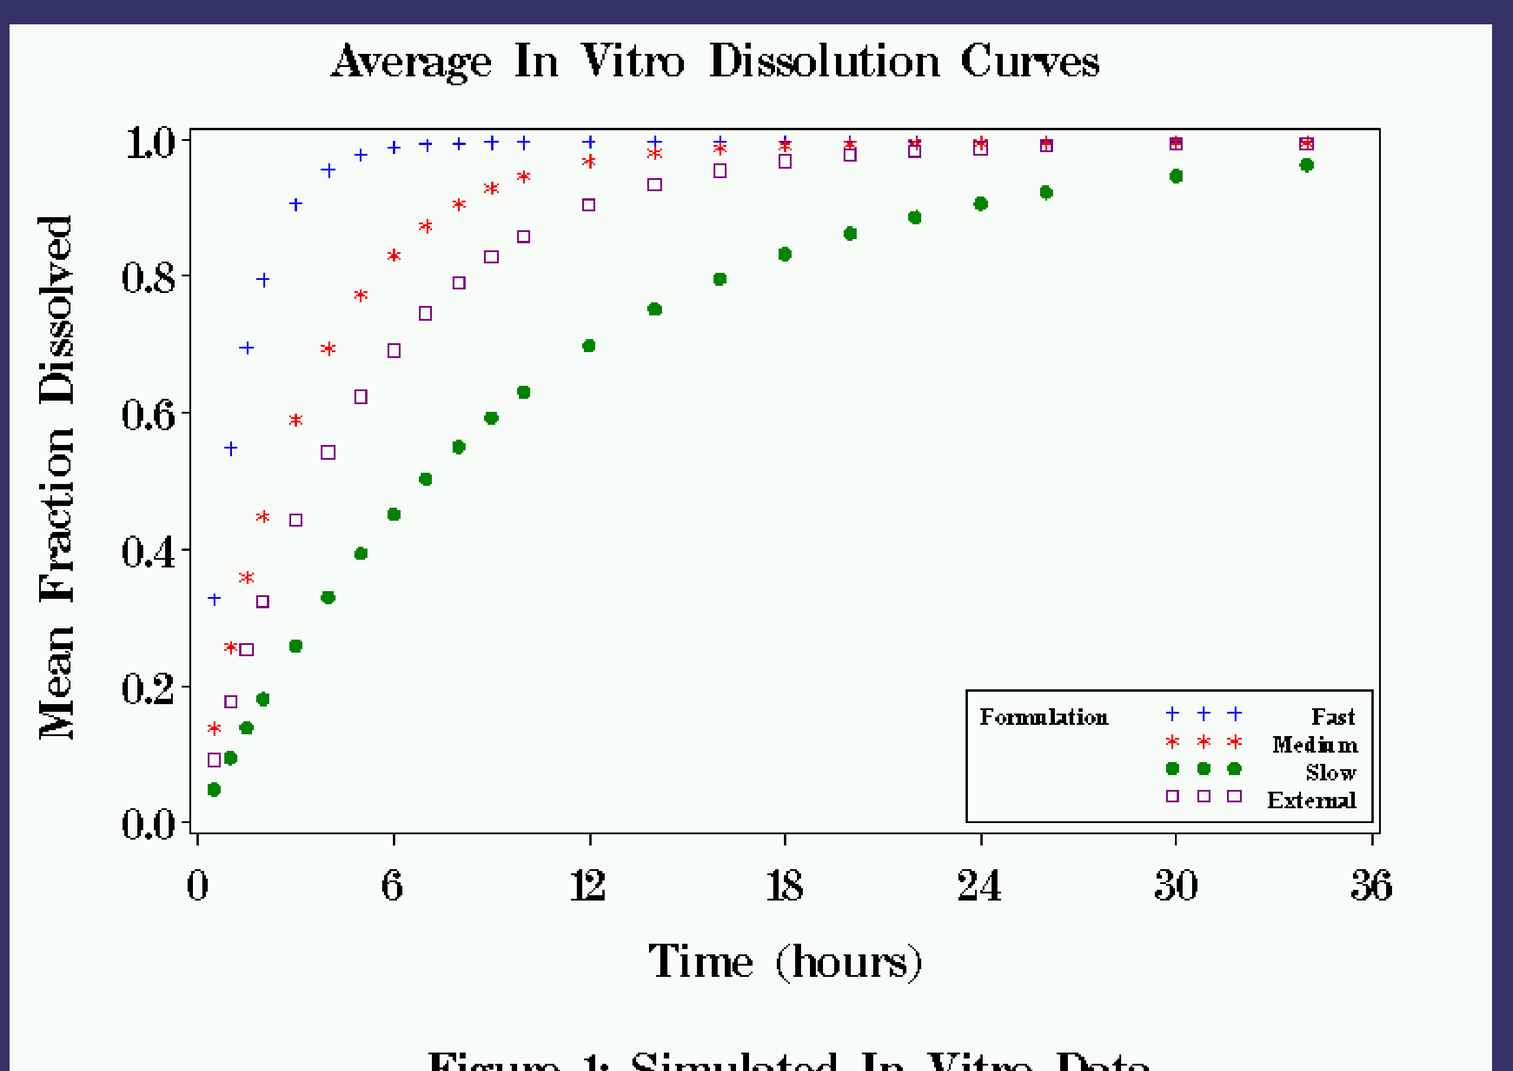

Average In Vitro Dissolution Curves
Mean Fraction Dissolved
Time (hours)
Formulation
Fast
Medium
Slow
External

Figure 1: Simulated In Vitro Data

The *in vivo* plasma concentration measured from the k^{th} subject following administration of the i^{th} tablet from formulation h is described as:

$$Y_{h2ik}(t) = Dose\ \phi_2 \int c_{\delta k}(t-\tau)\, F_{h2ik}'(\tau)\, d\tau + \varepsilon_{2ik}(t)$$
$$\varepsilon_{2ik}(t) \sim N(0, \sigma_2^2)$$

where $F_{h2ik}'(\tau)$ is the *in vivo* dissolution rate and ϕ_2 allows for a difference in bioavailability between the reference dose and the ER dose. The response of the k^{th} subject to a unit dose follows a standard one compartment pharmacokinetic model with first order absorption given by:

$$Y_{3k}(t) = c_{\delta k}(t) + \varepsilon_{3k}(t) \qquad \varepsilon_{3k}(t) \sim N(0, \sigma_3^2)$$
$$c_{\delta k}(t) = (\lambda_3/(\lambda_3-\lambda_2))\,(e^{-\lambda 2\,(t)} - e^{-\lambda 3\,(t)})$$

with λ_2 and λ_3 representing the rate constants of elimination and absorption of the drug respectively.

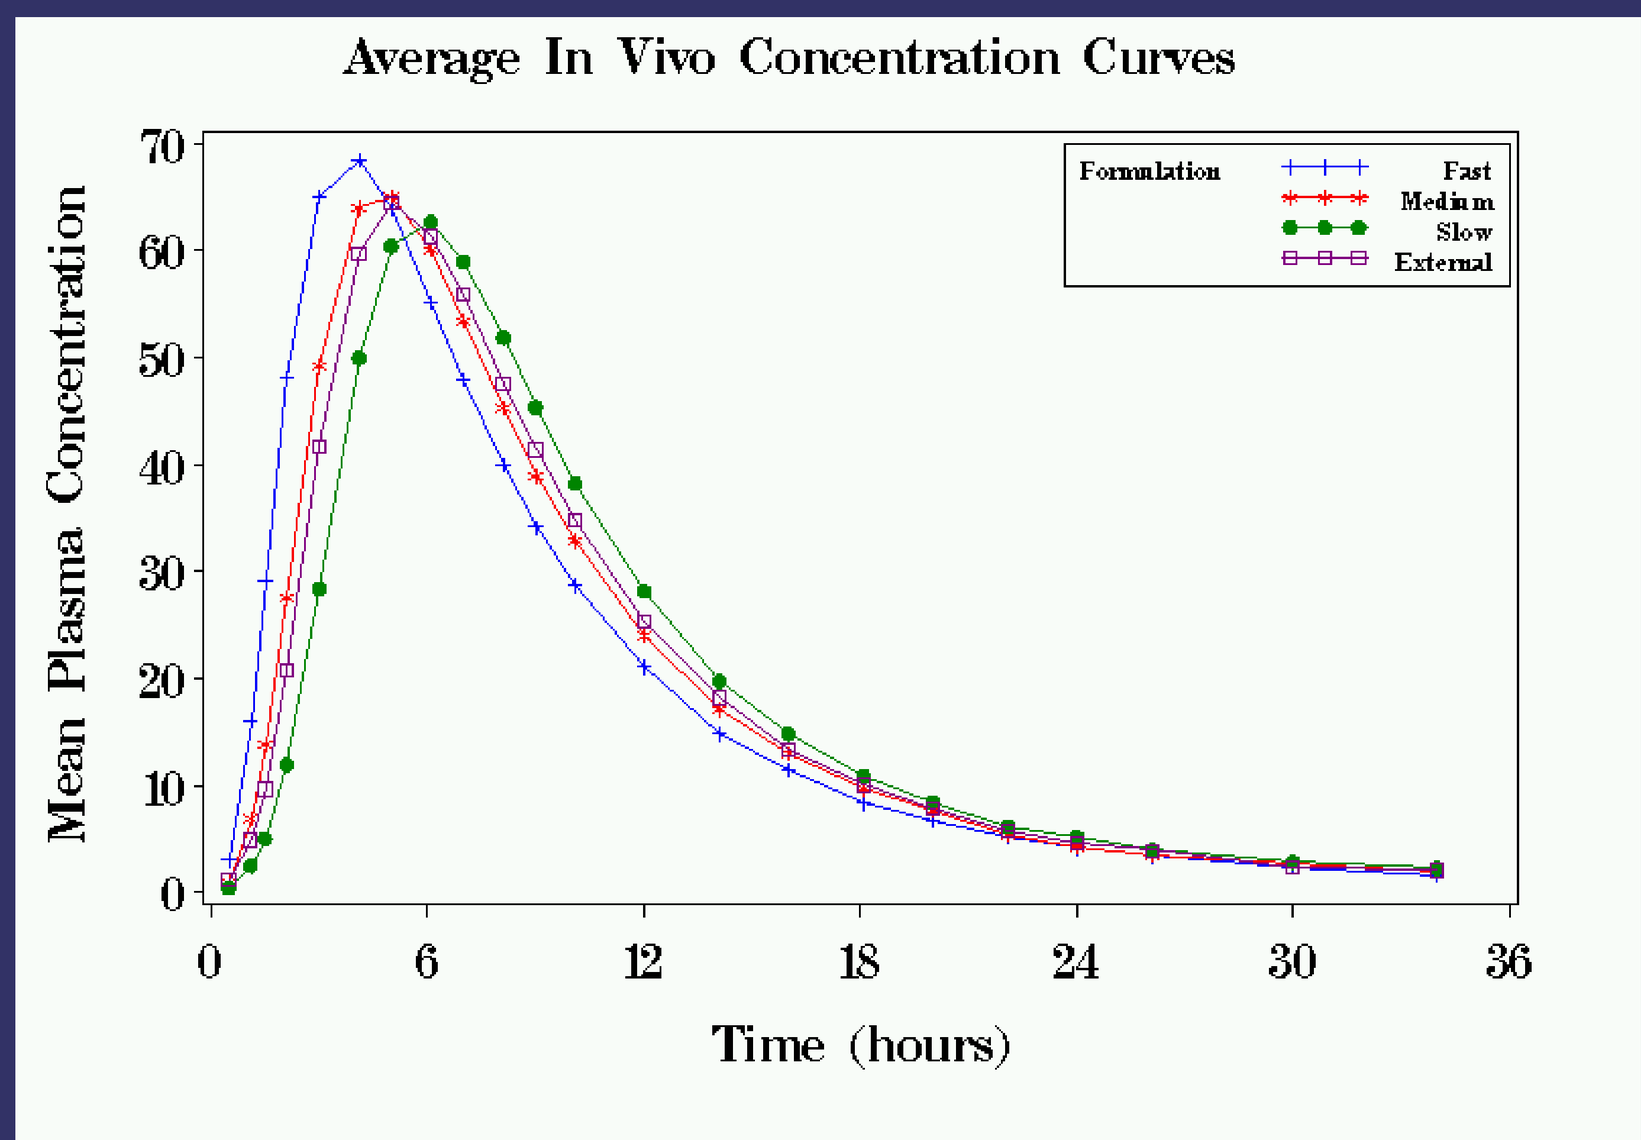

Average In Vivo Concentration Curves
Mean Plasma Concentration
Time (hours)
Formulation
Fast
Medium
Slow
External

Figure 2: Simulated In Vivo Data

The relationship between *in vitro* and *in vivo* dissolution is given by

$$logit\,(F_{h2ik}(t)) = logit\,(F_{h1i}(t)) + \theta_1 + u_i + s_{ik} + \theta_2 t$$
$$u_i \sim N(0, \omega_1^2)$$
$$s_{ik} \sim N(0, \omega_2^2)$$

where ω_2^2 gives the subject-to-subject variation.

In order to ensure that the results of this simulation could be meaningfully interpreted, the values of all the above parameters were based on those estimated using real datasets.

Each subject's reference data was analysed separately using the ADVAN2 subroutine provided in the NONMEM software developed by Beal and Sheiner [5]. Data from one formulation was excluded from the model-fitting stage to be used for the purposes of external validation. The remaining data was analysed twice to produce two predicted plasma concentration-time profiles. First the CoDe deconvolution method proposed by Hovorka et al [6, 7] was implemented. This was followed by the convolution based technique - a modified version of that reported by O'Hara et al [2] which implemented a custom written PRED subroutine for NONMEM. Each method's predictions were used to calculate the peak plasma concentration value and area under the plasma concentration curve for each formulation and were compared to the values calculated from the simulated data to calculate percentage prediction errors.

The US Food and Drug Administration (FDA) recommend assessment of prediction error for both the area under a plasma concentration curve (AUC) and for the peak plasma concentration (Cmax) when developing an IVIVC [4]. Predictions made using each method were used to compute the Cmax and AUC for each subject and compared to the known true values to calculate %PE.

RESULTS:

To illustrate the differences between the two modelling techniques, figure 3 shows simulated and predicted plasma concentration curves for each of the four batches averaged over all twelve subjects. The solid line represents predictions made when using the convolution method, while the dashed red line shows those of the deconvolution method.

The corresponding percentage prediction errors for each formulation resulting from analysis using each method are shown in table I.

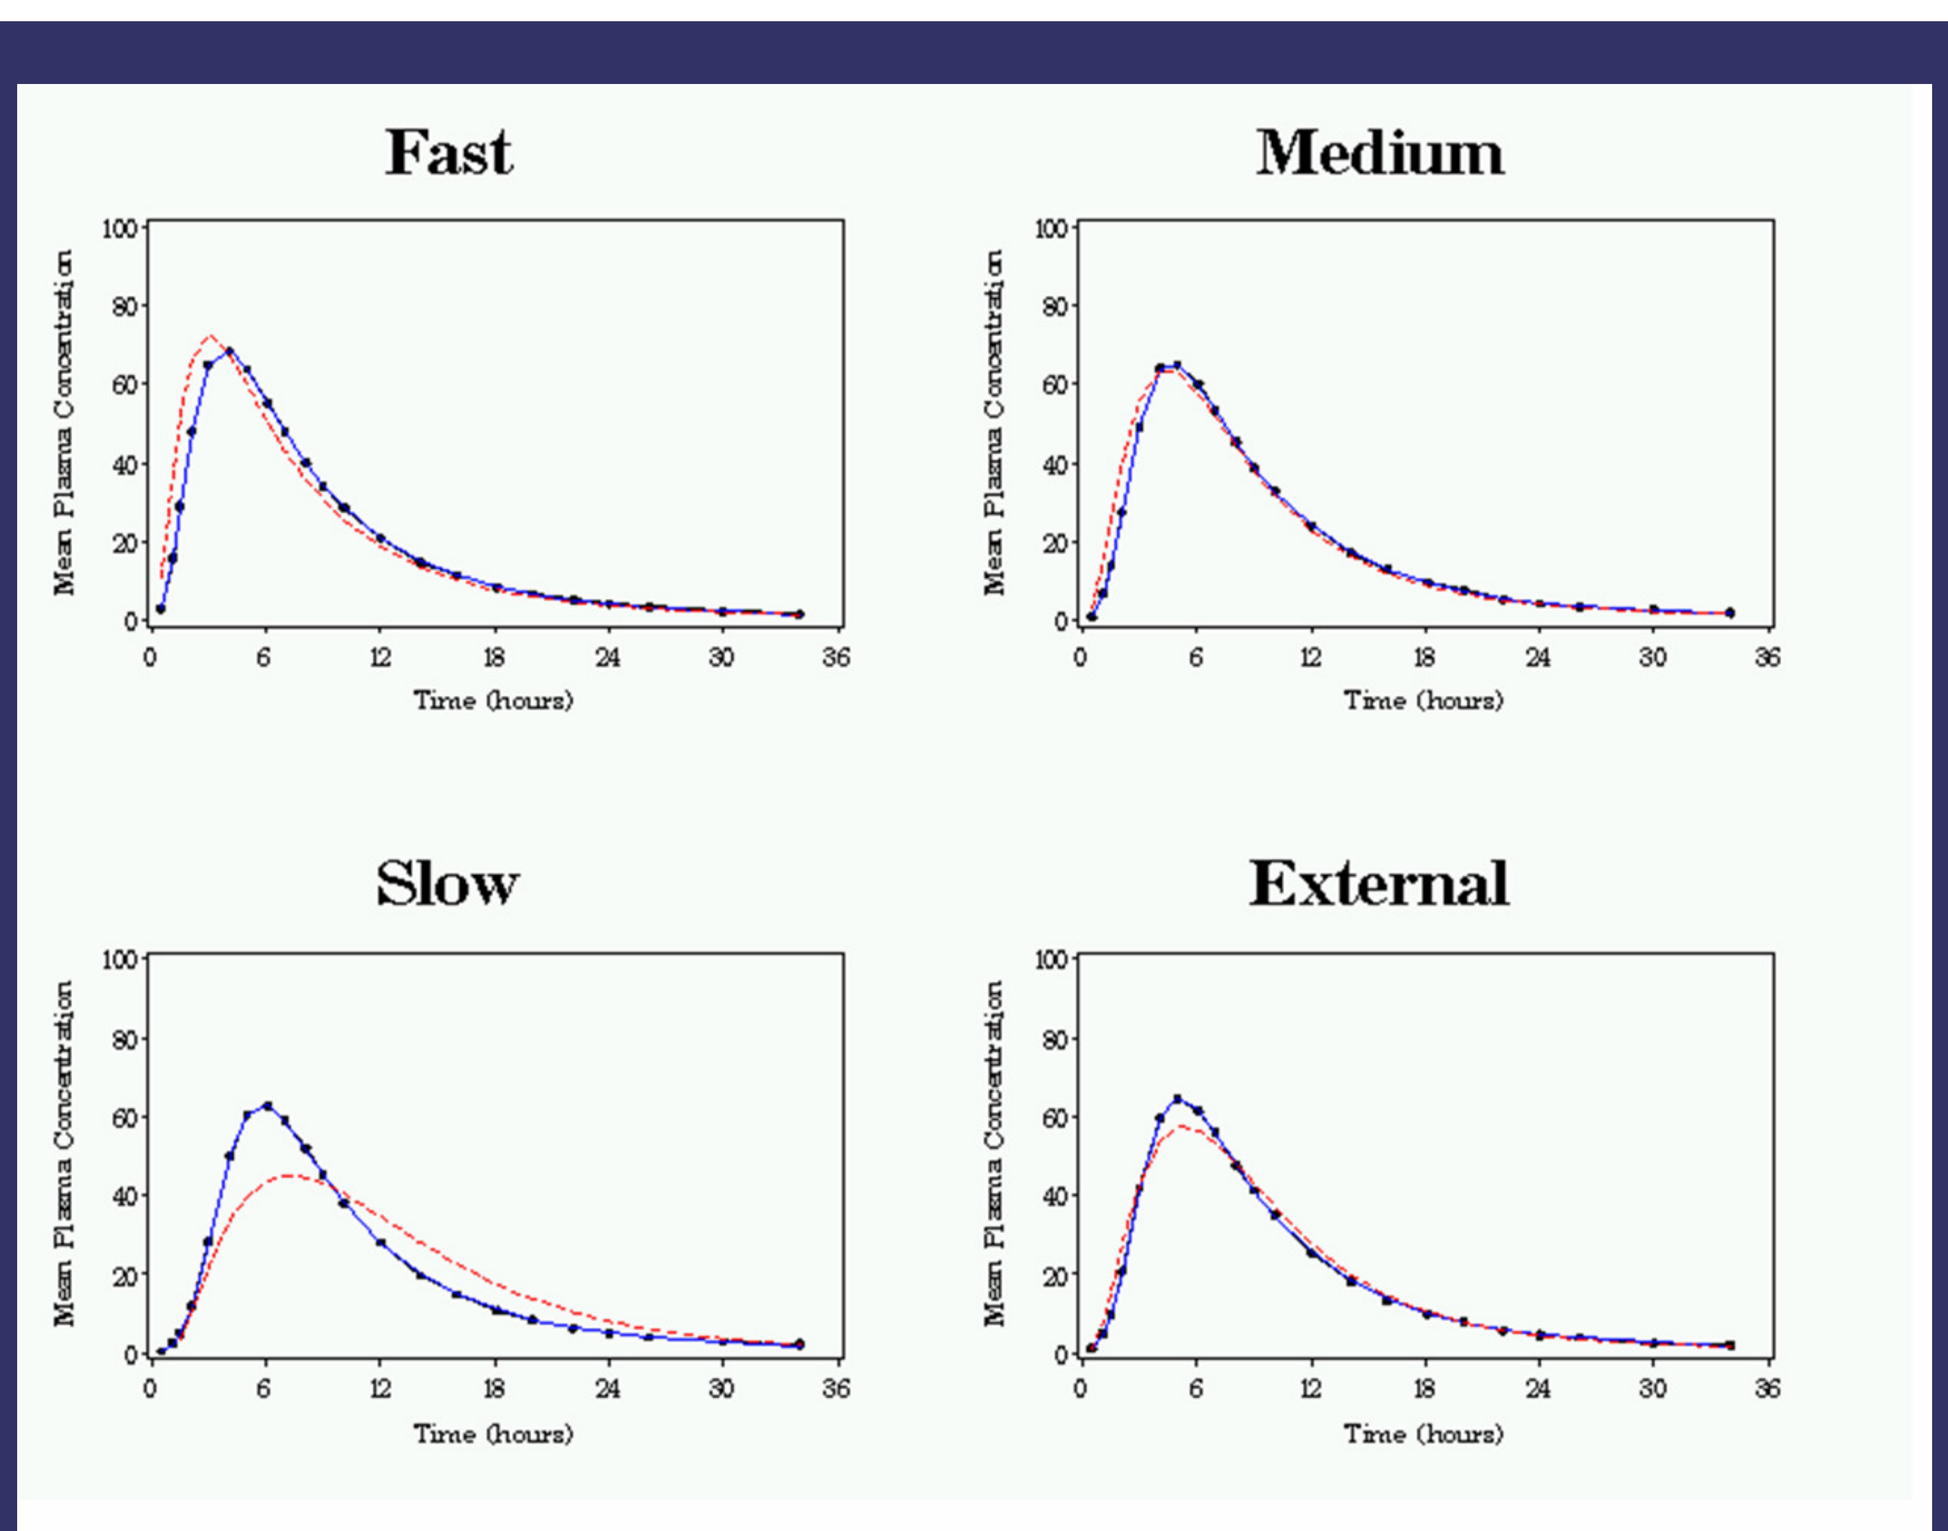

Fast
Medium
Slow
External
Mean Plasma Concentration
Time (hours)

Figure 3: Observed and Predicted Plasma Concentration Curves

BATCH	METHOD	%PE AUC	%PE CMAX
FAST	DECONVOLUTION	-0.47	-6.14
	CONVOLUTION	-0.24	-0.03
MEDIUM	DECONVOLUTION	-0.2	2.23
	CONVOLUTION	0.04	-0.37
SLOW	DECONVOLUTION	-0.57	28.04
	CONVOLUTION	-0.7	0.03
EXTERNAL	DECONVOLUTION	-0.27	11.03
	CONVOLUTION	0.002	0.02

Table I: Summary of %PE Results

DISCUSSION AND CONCLUSIONS:

The development of *In Vitro – In Vivo* Correlation models is an important step in drug development and, as with all aspects of this process, precision is vital. The method most frequently employed at present, i.e. the conventional deconvolution based method, is statistically flawed and performs inadequately, especially by comparison to the alternative non linear mixed effects modelling technique. The findings of this study confirm the expectation that the convolution based method would outperform its counterpart.

The concerns about the deconvolution method are supported by the results and its lack of accuracy in prediction is a result of the combined influence of its theoretical flaws. While these may have seemed like purely statistical considerations, they have a very striking practical effect in terms of quality of predictions and, therefore, ability to meet the FDA validation criteria.

The fact that the conventional approach frequently fails to establish an IVIVC when it really does exist (i.e. fails the FDA test when it ought to pass) should be of great concern to those currently implementing this method. These results substantiate the statistical theory, illustrating and quantifying the superiority of the convolution based approach and providing support for its use in preference to the deconvolution-based group of methods.

REFERENCES:

[1] Dunne A., Gaynor C. and Davis J. (2005) Deconvolution Based Approach for Level A In Vivo-In Vitro Correlation Modelling: Statistical Considerations. *Clinical Research and Regulatory Affairs*, **22**, 1-14.

[2] O'Hara T., Hayes S., Davis J., Devane J., Smart T. and Dunne A. (2001) In Vivo-In Vitro Correlation (IVIVC) Modeling Incorporating a Convolution Step. *Journal of Pharmacokinetics and Pharmacodynamics*, **28**, 277-298.

[3] Gaynor, C. Dunne, A.and Davis, J. (2006) Comparison of conventional In Vitro - In Vivo Correlation methodology with non-linear mixed effects modelling. *PAGE 15 2006* [www.page-meeting.org/?abstract=953]

[4] Food and Drug Administration (1997) Guidance for Industry: Extended Release Oral Dosage Forms: Development, Evaluation, and Application of In Vitro/In Vivo Correlations.

[5] S.L. Beal and L. B. Sheiner. *NONMEM User's Guides, NONMEM Project Group,* University of California, San Francisco, 1992.

[6] Hovorka, R., Chappell, M.J., Godfrey, K.R., Madden, F.N., Rouse, M.K. and Soons, P.A. (1998) CODE: A Deconvolution Program Implementing a Regularization Method of Deconvolution Constrained to Non-Negative Values. Description and Pilot Evaluation. *Biopharmaceutics and Drug Disposition* **19**, 39-53.

[7] Madden, F. N., Godfrey, K.R., Chappell, M.J., Hovorka, R. and Bates, R. A. (1996) A Comparison of Six Deconvolution Techniques. *Journal of Pharmacokinetic and Biopharmaceutics,* **24**: 3, 283-299.